Long-term safety and tolerability of STS101, a novel investigational dihydroergotamine nasal powder: initial data from the ASCEND study

Stewart J. Tepper, MD[1], Shannon Strom, PhD[2], Detlef Albrecht, MD[2]

[1]Geisel School of Medicine at Dartmouth and Dartmouth-Hitchcock Medical Center, Lebanon, NH, USA; [2]Satsuma Pharmaceuticals, Inc., South San Francisco, CA, USA

Introduction

- STS101, a novel investigational dihydroergotamine mesylate (DHE) nasal powder formulation delivered via an easy-to-use, easy-to-carry, pre-filled single-use device, is designed for intranasal administration for the acute treatment of migraine (with or without aura).
- Common side effects of DHE include nausea and vomiting (regardless of dosage form), which have been attributed to high peak plasma concentrations after initial administration.[1,2]
- While liquid nasal sprays have lower reported occurrences of nausea and vomiting, irritative nasal symptoms are common.[1,2]
- Two previous Phase 1 studies of STS101 showed that a dose level of 5.2 mg rapidly achieved plasma concentrations in the target therapeutic range (approaching those of intramuscular DHE and 2-3-fold higher than liquid nasal sprays) with a favorable tolerability profile in healthy subjects.[3,4]

Objective

- To report preliminary safety and tolerability data from an ongoing, long-term (12 month), open-label Phase 3 study for STS101 5.2 mg for the acute treatment of migraine attacks.

Methods

Study design and treatment intervention

- ASCEND (NCT04406649) is an ongoing, open-label, 12 month study of STS101 (DHE nasal powder) in adults aged 18–65 years with migraine.
 - The data cutoff date for this preliminary analysis was June 30, 2021 and includes adverse events reported up to 9 months of study drug exposure in some subjects.
- After establishing eligibility, subjects could self-administer STS101 5.2 mg up to 2 doses within 24 hours to treat a single migraine attack, and up to 12 doses per month (Figure 1).

Subjects

- Study subjects must have ≥1-year history of migraine (with or without aura) according to the International Classification of Headache Disorders, 3rd edition, including[5]:
 - Migraine onset before age of 50 years
 - 4–12 migraine attacks/month in each of the 3 months prior to screening
 - <15 headache days/month in each of the 3 months prior to screening
- Exclusion criteria included subjects with a diagnosis of non-migraine headache, history of cerebrovascular disease, and those with ≥2 cardiovascular risk factors.
- Subjects must have an intact nasal mucosa at baseline (i.e., no ulceration or bleeding; no or mild erythema, swelling, and rhinorrhea).

Outcomes and Analyses

- Safety evaluations include physical and nasal examinations, vital signs, ECGs, laboratory tests, and treatment emergent adverse event (TEAE) assessments.
- TEAE assessment will be performed at every study visit (months 1–6, 8, 10, and 12).
- Blood pressure and assessment of nasal symptoms will be evaluated at every visit (screening, baseline, and months 1–6, 8, 10 and 12).
- A 12-lead ECG will be performed in triplicate at screening, baseline, and months 3, 6, 8, 10, and 12.

Results

Subjects

- The safety population included 273 subjects who treated 4247 migraine attacks, with 143 completing 6 months of treatment while treating 3653 migraine attacks (average of 4.3 treated attacks per month).
 - Mean age: 39 ± 11 years
 - 89% female
 - 84% Caucasian (36% Hispanic)
- Of the total migraine attacks, 670 (15.8%) were treated with a second dose for a total of 4917 STS101 doses used.

Discontinuation due to adverse events

- A total of 20 (7.3%) subjects discontinued the study due to TEAEs, which were deemed treatment-related in 18 subjects (Table 1).
- Of the 2 serious adverse events (SAEs) reported (postural orthostatic tachycardia syndrome, cholecystitis), neither were treatment-related.

Adverse events

- In total, at least one treatment-related TEAE was observed in 31.1% (n=85) of subjects, with most frequent being nasal discomfort (13.9%), dysgeusia (7.7%), and nasal congestion (5.5%) (Table 2).
- Treatment-related TEAEs were observed in 6.4% of migraine attacks (n=273/4247).
- TEAEs were generally mild and transient, with no treatment-related SAEs.
- Among nasal TEAEs observed, 95% were assessed as mild.

TEAEs by version of delivery device

- Introduction of an optimized STS101 intranasal delivery device during the study did not result in any observed differences in TEAE rates or severity between the original and optimized delivery devices (Table 3).

Nasal examinations

- No nasal findings were observed for the majority of nasal assessments at baseline and 6 months with the exception of one instance of nasal edema, and one of rhinorrhea, both of moderate severity, which were reported at 6 months (Table 4).
- No clinically relevant changes in ECGs or blood pressure evaluations were observed from baseline to 6 months (Table 5).

Figure 1. STS101 administration

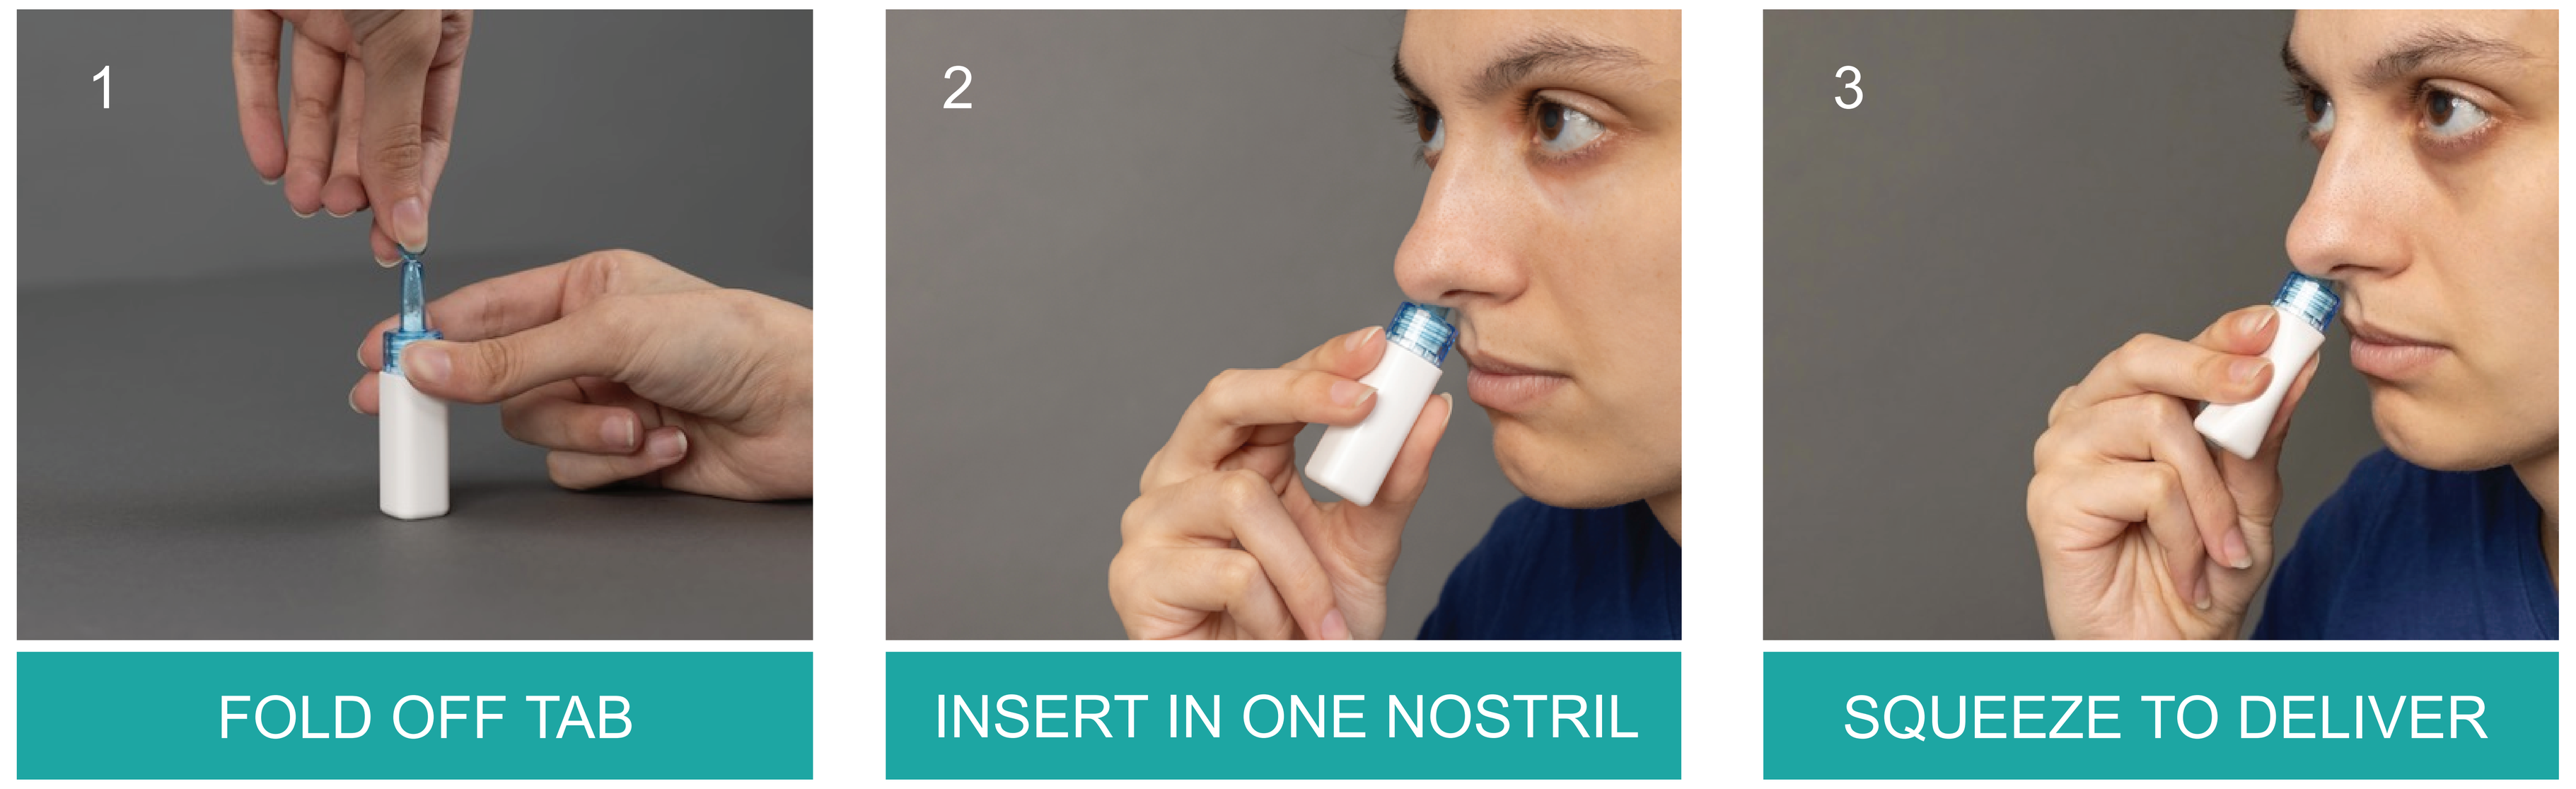
1
FOLD OFF TAB
2
INSERT IN ONE NOSTRIL
3
SQUEEZE TO DELIVER

Table 1. Treatment discontinuations due to TEAEs

	Subjects N=273
Any TEAE leading to discontinuation, n (%)	20 (7.3%)
Treatment-related TEAEs leading to discontinuation, n (%)	18 (6.6%)
Nasal burning, discomfort, or other nasal AEs	8 (2.9%)
Vomiting/nausea	3 (1.1%)
Worsening of migraine	2 (0.7%)
Leg pain	2 (0.7%)
Abdominal pain	1 (0.4%)
Face pain	1 (0.4%)
Upper gum pain	1 (0.4%)

AE, adverse event; TEAE, treatment-emergent adverse event.

Table 2. Summary of reported TEAEs

	Total Subjects N=273	Total Migraine Attacks N=4247
	n (%) with ≥1 TEAE	n (%) with ≥1 TEAE
Any TEAE	132 (48.4%)	451 (10.6%)
Treatment-related TEAE	85 (31.1%)	273 (6.4%)
Most frequent treatment-related TEAEs	**n (%) reporting TEAE at least once**	**n (%) attacks with TEAE**
Nasal discomfort	38 (13.9%)	90 (2.1%)
Dysgeusia	21 (7.7%)	119 (2.8%)
Nasal congestion	15 (5.5%)	46 (1.1%)
Nausea	10 (3.7%)	10 (0.2%)
Rhinorrhea	8 (2.9%)	13 (0.3%)
Vomiting	8 (2.9%)	6 (0.1%)
Epistaxis	5 (1.8%)	6 (0.1%)
Lacrimation increased	4 (1.5%)	14 (0.3%)

TEAE, treatment-emergent adverse event.

References

1. Ziegler DK, et al. *Neurology.* 1994;44(3).
2. Silberstein SD, et al. *Headache.* 2020;60(1):40-57.
3. Albrecht D, et al. *Headache.* 2020;60(4):701-712.
4. Safety and tolerability results based on November 9, 2021 STS101-006 Study.
5. Headache Classification Committee of the International Headache Society (IHS). The International Classification of Headache Disorders, 3rd edition. *Cephalalgia.* 2018;38(1):1-211.

Disclosures

Dr. Tepper received research grants (no personal compensation) from Allergan/Abbvie, Amgen, Eli Lilly, Lundbeck, Neurolief, Novartis, Satsuma, Zosano; is a consultant and/or advisory board bember (honoraria) for Aeon, Allergan/Abbvie, Alphasights, Amgen, Aruene, Atheneum, Axsome Therapeutics, Becker Pharmaceutical Consulting, BioDelivery Sciences International, Biohaven, ClearView Healthcare Partners, CoolTech, CRG, Currax, Decision Resources, Defined Health, DRG, Eli Lilly, ExpertConnect, FCB Health, Fenix, GLG, Guidepoint Global, Health Advances, Health Science Communications, HMP Communications, Impel, InteractiveForums, Keyquest, Krog and Partners, Lundbeck, M3 Global Research, Magnolia Innovation, MJH Holdings, Neurolief, Novartis, P Value Communications, Pain Insights, Inc, Palion Medical, Pulmatrix, Putnam Associates, SAI MedPartners, Satsuma, Slingshot Insights, Spherix Global Insights, Strategy Inc, System Analytic, Taylor and Francis, Teva, Theranica, Tremeau, Trinity Partners, Unity HA, XOC, Zosano; receives salary from Dartmouth-Hitchcock Medical Center, Thomas Jefferson University; received CME honoraria from American Academy of Neurology, American Headache Society, Annenberg Center for Health Sciences, Catamount Medical Education, Diamond Headache Clinic, Forefront Collaborative, Haymarket Medical Education, Medical Education Speakers Network, Medical Learning Institute Peerview, Migraine Association of Ireland, Miller Medical Education, National Association for Continuing Education, North American Center for CME, The Ohio State University, Physicians' Education Resource, PlatformQ Education, Primed, Texas Neurological Society, Vindico Medical Education, WebMD/Medscape

Drs. Strom and *Albrecht* are employees of Satsuma Pharmaceuticals.

Acknowledgements

The authors also thank The Medicine Group, LLC (New Hope, PA, USA) for providing medical writing support, which was funded by Satsuma Pharmaceuticals, Inc. (South San Francisco, CA, USA) in accordance with Good Publication Practice guidelines.

Table 3. Summary of TEAEs

	1st-Generation device	2nd-Generation device
Total attacks treated, N	1939	2290
Attacks with ≥1 TEAE, n (%)	283 (6.7%)	274 (6.5%)
Most frequent TEAEs	**% of Attacks**	
Nasal discomfort	4.7%	4.1%
Dysgeusia	5.8%	5.1%
Nasal congestion	2.6%	2.1%
Nausea	0.8%	0.7%
Rhinorrhea	0.8%	0.4%
Nasal obstruction	0.9%	0.7%
Vomiting	0.5%	0.4%
Epistaxis	0.3%	0.2%
Lacrimation increased	0.7%	0.3%

TEAE, treatment-emergent adverse event.

Table 4. Summary of objective nasal assessment of symptoms (N=143)

Subjects, n (%)	Month 6	Baseline		
		None	Mild	Moderate
Nasal bleeding	None	142 (99.3%)	1 (0.7%)	0
	Mild	0	0	0
	Moderate	0	0	0
Nasal edema	None	137 (95.8%)	3 (2.1%)	1 (0.7%)
	Mild	2 (1.4%)	0	0
	Moderate	0	0	0
Nasal erythema	None	137 (95.8%)	5 (3.5%)	0
	Mild	0	0	0
	Moderate	1 (0.7%)	0	0
Nasal ulceration	None	142 (100%)	0	0
	Mild	0	0	0
	Moderate	0	0	0
Rhinorrhea	None	137 (95.8%)	4 (2.8%)	1 (0.7%)
	Mild	1 (0.7%)	0	0
	Moderate	0	0	0

Table 5. Summary of ECG and blood pressure evaluations

	Baseline mean ± SD	6 Months mean ± SD
Heart rate (beats/min)	68.9 ± 10.26	69.4 ± 11.09
RR (msec)	883.6 ± 130.28	879.2 ± 132.11
PR (msec)	153.9 ± 19.73	155.0 ± 18.41
QRS (msec)	88.3 ± 9.66	87.4 ± 11.47
QTcF (msec)	407.9 ± 17.59	407.7 ± 22.47
Diastolic blood pressure	74.6 ± 8.07	75.1 ± 7.76
Systolic blood pressure	116.3 ± 10.31	116.2 ± 10.34

Conclusions

- STS101 (an investigational DHE nasal powder) was well tolerated by subjects with migraine when used long-term and on a PRN (as needed) basis during the ASCEND study.
- Incidence of discontinuations due to treatment-related TEAEs was low (6.6%).
- The use of the two versions of the STS101 delivery device did not result in differences in incidence rates or severity for the most common TEAEs (nasal discomfort, dysgeusia, and nasal congestion).
- Nasal evaluations, ECGs, and blood pressure assessments did not identify any safety signals.

Presented at the American Academy of Neurology (AAN) 74th Annual Meeting; April 2–7, 2022 (Seattle, WA), April 24-26, 2022 (Virtual)