Relationship of Dihydroergotamine (DHE) Pharmacokinetic Parameters and Clinical Efficacy and Systemic Side Effects

P-193

John Kollins, BSE, MBA; Shannon Strom, PhD; and Detlef Albrecht, MD

Satsuma Pharmaceuticals Inc., South San Francisco, CA, United States

Introduction

- Dihydroergotamine mesylate (DHE) exerts anti-migraine effects via a unique multi-modal mechanism of action involving interactions with both serotonergic and adrenergic receptors, has been used since 1946 for the acute treatment of migraine, and is recognized as a first-line treatment option.[1-4]
- DHE is available as a sterile injectable solution for subcutaneous (SC), intramuscular (IM), or intravenous (IV) administration, and as a liquid nasal spray (LNS).[5-7]
- Different DHE dosage forms having dramatically different pharmacokinetic (PK) and adverse event (AE) profiles are commercially available and/or have been evaluated in large, randomized controlled trials.
- However, the relationships among PK, clinical efficacy, and the signature DHE side effects of nausea/vomiting—which have been reported to be C_{max} dependent[1,8]—have not been comprehensively evaluated.
- STS101 is a novel investigational DHE product designed to consistently achieve a PK profile that maximizes efficacy while minimizing side effects, in particular nausea and vomiting; STS101 comprises a DHE powder formulation and an easy-to-use, easy-to-carry, single-use, disposable intranasal delivery device designed to facilitate quick and intuitive self-administration into a single nostril with no assembly or priming required (Figure 1).

Objective

- To further define the relationship among DHE PK parameters, clinical efficacy, and tolerability.

Methods

Determination of relationships between DHE PK and clinical efficacy

- A literature search was performed to identify randomized, controlled clinical trials (RCTs) of various DHE dosage forms administered single-dose in which clinical efficacy (2-hour pain relief [2hPR]) was reported.
- For each DHE dosage form for which a qualifying RCT was identified, PK parameters (mean C_{max}, $AUC_{0\text{-}0.5h}$, $AUC_{0\text{-}1h}$, $AUC_{0\text{-}2h}$, T_{max}) of that dosage form were determined from PK studies previously reported in the published literature. (Note: Reported partial AUC values were used when available; when partial AUC values were not reported, computer-based graphical analysis of digitized PK curves from the published literature was performed, and the trapezoidal rule was applied to calculate partial AUC values.[10] In addition, to the extent possible, PK results from trials employing modern analytical methods [liquid chromatography, tandem mass spectrometry] for determining DHE plasma concentrations were utilized.)
- For each PK parameter, and across all DHE dosage forms, univariate linear regression analyses using the ordinary least squares method were performed to determine correlations with clinical efficacy (2hPR) based on the calculated coefficients of determination (R-squared [R^2]). Sensitivity analyses (not shown) were performed to confirm robustness of the relationships between PK parameters and 2hPR.

Determination of relationships between DHE C_{max} and nausea

- Mean DHE C_{max} and nausea frequency data from a Phase 1 clinical trial in which a total of 35 healthy volunteers (aged 18–50 years) were administered single doses of 2.0 mg LNS DHE, 1.0 mg IM DHE, and three different dose strengths of STS101 DHE nasal powder (5.2 mg, 7.0 mg, and 8.6 mg) in a 5-period, cross-over design were analyzed.[9]
 – These data were compared with literature reports of Phase 1 DHE studies in which no anti-emetics were administered and mean DHE C_{max} values and nausea frequency were reported for various formulations and administration routes

Results

Relationships between DHE PK and clinical efficacy

- The literature search identified 9 RCTs (4 undertaken with LNS DHE, 3 with two different doses and formulations of DHE administered via SC injection, and 2 with oral pulmonary [OP] DHE) in which 2hPR was reported (Table 1).
- PK parameters (mean C_{max}, T_{max}, and drug exposure at 0.5h, 1h, and 2h post-dose expressed as the corresponding partial area-under-the-curve [AUC]) for the DHE dosage forms were as described in Table 2.
- The coefficient of determination (R^2) values calculated to evaluate the degree of correlation between each PK parameter and the 2hPR rates reported for the RCTs were as shown in Table 3.
 – DHE C_{max} and $AUC_{0\text{-}0.5h}$ showed the highest R^2 values and thus were the best PK predictors of clinical efficacy at 2h
 – C_{max} and $AUC_{0\text{-}0.5h}$ R^2 values increased to 0.8 if the LNS DHE outlier[11] was excluded (Table 3 and Figures 2–3)

Relationship between DHE C_{max} and nausea

- In a Phase 1 study in which 35 healthy subjects were administered LNS DHE, IM DHE, and STS101, increased rates of nausea were observed with increasing mean C_{max} and particularly mean C_{max} concentrations exceeding approximately 2500 pg/mL (Table 4).
- Results from Phase 1 studies of various DHE dosage forms (LNS, IM, OP, IV and nasal powder/STS101) administered to healthy volunteers indicate nausea incidence was minimal when mean C_{max} concentrations were below approximately 2500 pg/mL; above this threshold C_{max} concentration, nausea incidence increased as C_{max} increased (Figure 4).

Figure 1. STS101 Administration

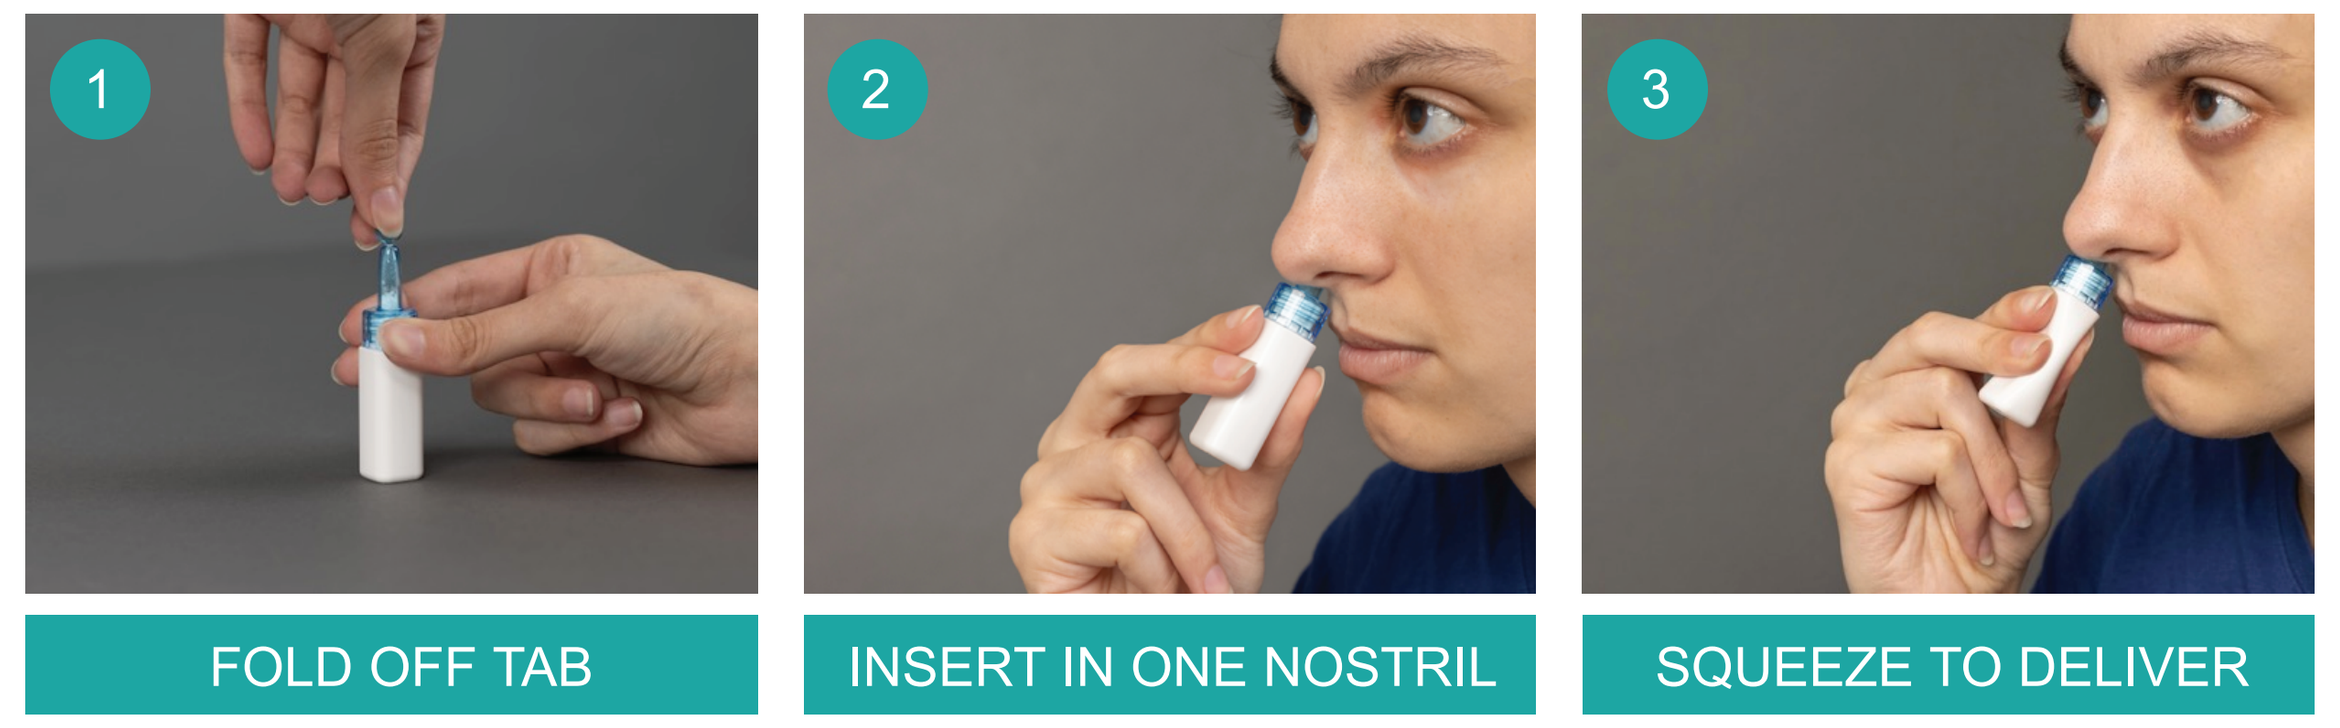
1
FOLD OFF TAB
2
INSERT IN ONE NOSTRIL
3
SQUEEZE TO DELIVER

Table 1. Placebo/Active Controlled Phase 3 Efficacy Studies Included in Pharmacokinetic/Pharmacodynamic Efficacy Analysis

DHE Route of Administration / Dosage Form	Subject Demographics (age; years of migraine; race)	2-Hour Pain Relief (%)
Nasal; liquid spray; 2.0 mg DHE *(Valeant, Migranal Prescribing Information 2017)*	Age: 39 years Years migraine: NR White: 95%	Study 1: 61 Study 2: 47* Study 3: 32* Study 4: 30*
SC injection; solution; 1.0 mg DHE (MT300) *(MT 300 — Pozen. Drugs in R & D. 2003)*	NR	Study 1: 51 Study 2: 48
Pulmonary; powder; 1.0 mg DHE (MAP0004) *(Aurora et al., Headache. 2009; Aurora et al., Headache. 2011)*	Age: 39.8; 40.5 years Years migraine: 21.3; NR White: 84.0%; 88.1%	Study 1: 73 Study 2: 59
SC injection; solution; 1.0 mg DHE *(Winner et al., Arch Neurol. 1996)*	Age: 40.5 years Years migraine: NR White: NR	73

*Not statistically significant different vs placebo.
DHE, dihydroergotamine; NR, not reported; SC, subcutaneous.

Table 2. Phase 1 Pharmacokinetic Studies Included in Pharmacokinetic/Efficacy Analysis

DHE Route of Administration / Dosage Form	C_{max} (pg/mL)	$AUC_{0\text{-}0.5h}$ (h*pg/mL)	$AUC_{0\text{-}1h}$ (h*pg/mL)	$AUC_{0\text{-}2h}$ (h*pg/mL)	T_{max} (median; min)
Nasal; liquid spray; 2.0 mg DHE *(Albrecht et al., AAN 2022 Annual Meeting Abstracts; Albrecht et al., Headache. 2020; Shrewsbury et al., Headache. 2019**)*	645*	102*	384*	875*	56*
SC injection; solution; 1.0 mg DHE (MT300) *(Plachetka, John R., and Donna Gilbert. U.S. Patent No. 7,060,694. 2006)*	1178**	496**	906**	2035**	NR
Pulmonary; powder; 1.0 mg DHE (MAP0004) *(Kellerman et al., J Aerosol Med Pulm Drug Deliv. 2013)*	2720	810**	1343**	1447	9
SC injection; solution; 1.0 mg DHE *(Schran et al., Curr Ther Res. 1994)*	3210	1174**	2348**	4003**	22

*Mean from three Phase 1 trials. **From computer-based graphical analysis.
DHE, dihydroergotamine; NR, not reported; SC, subcutaneous.

Table 3. R^2 Values Calculated for Pharmacokinetic/Efficacy Analysis

Statistic	C_{max}	$AUC_{0\text{-}0.5h}$	$AUC_{0\text{-}1h}$	$AUC_{0\text{-}2h}$	T_{max}
R-squared value (PK parameter versus 2hPR)	0.62	0.60	0.57	0.33	0.36
R-squared value (PK parameter versus 2hPR) *Excluding LNS DHE (Migranal) outlier from analysis[11]*	0.83	0.84	0.77	0.44	0.57

Figure 2. Relationship Between DHE C_{max} and 2-Hour Pain Relief

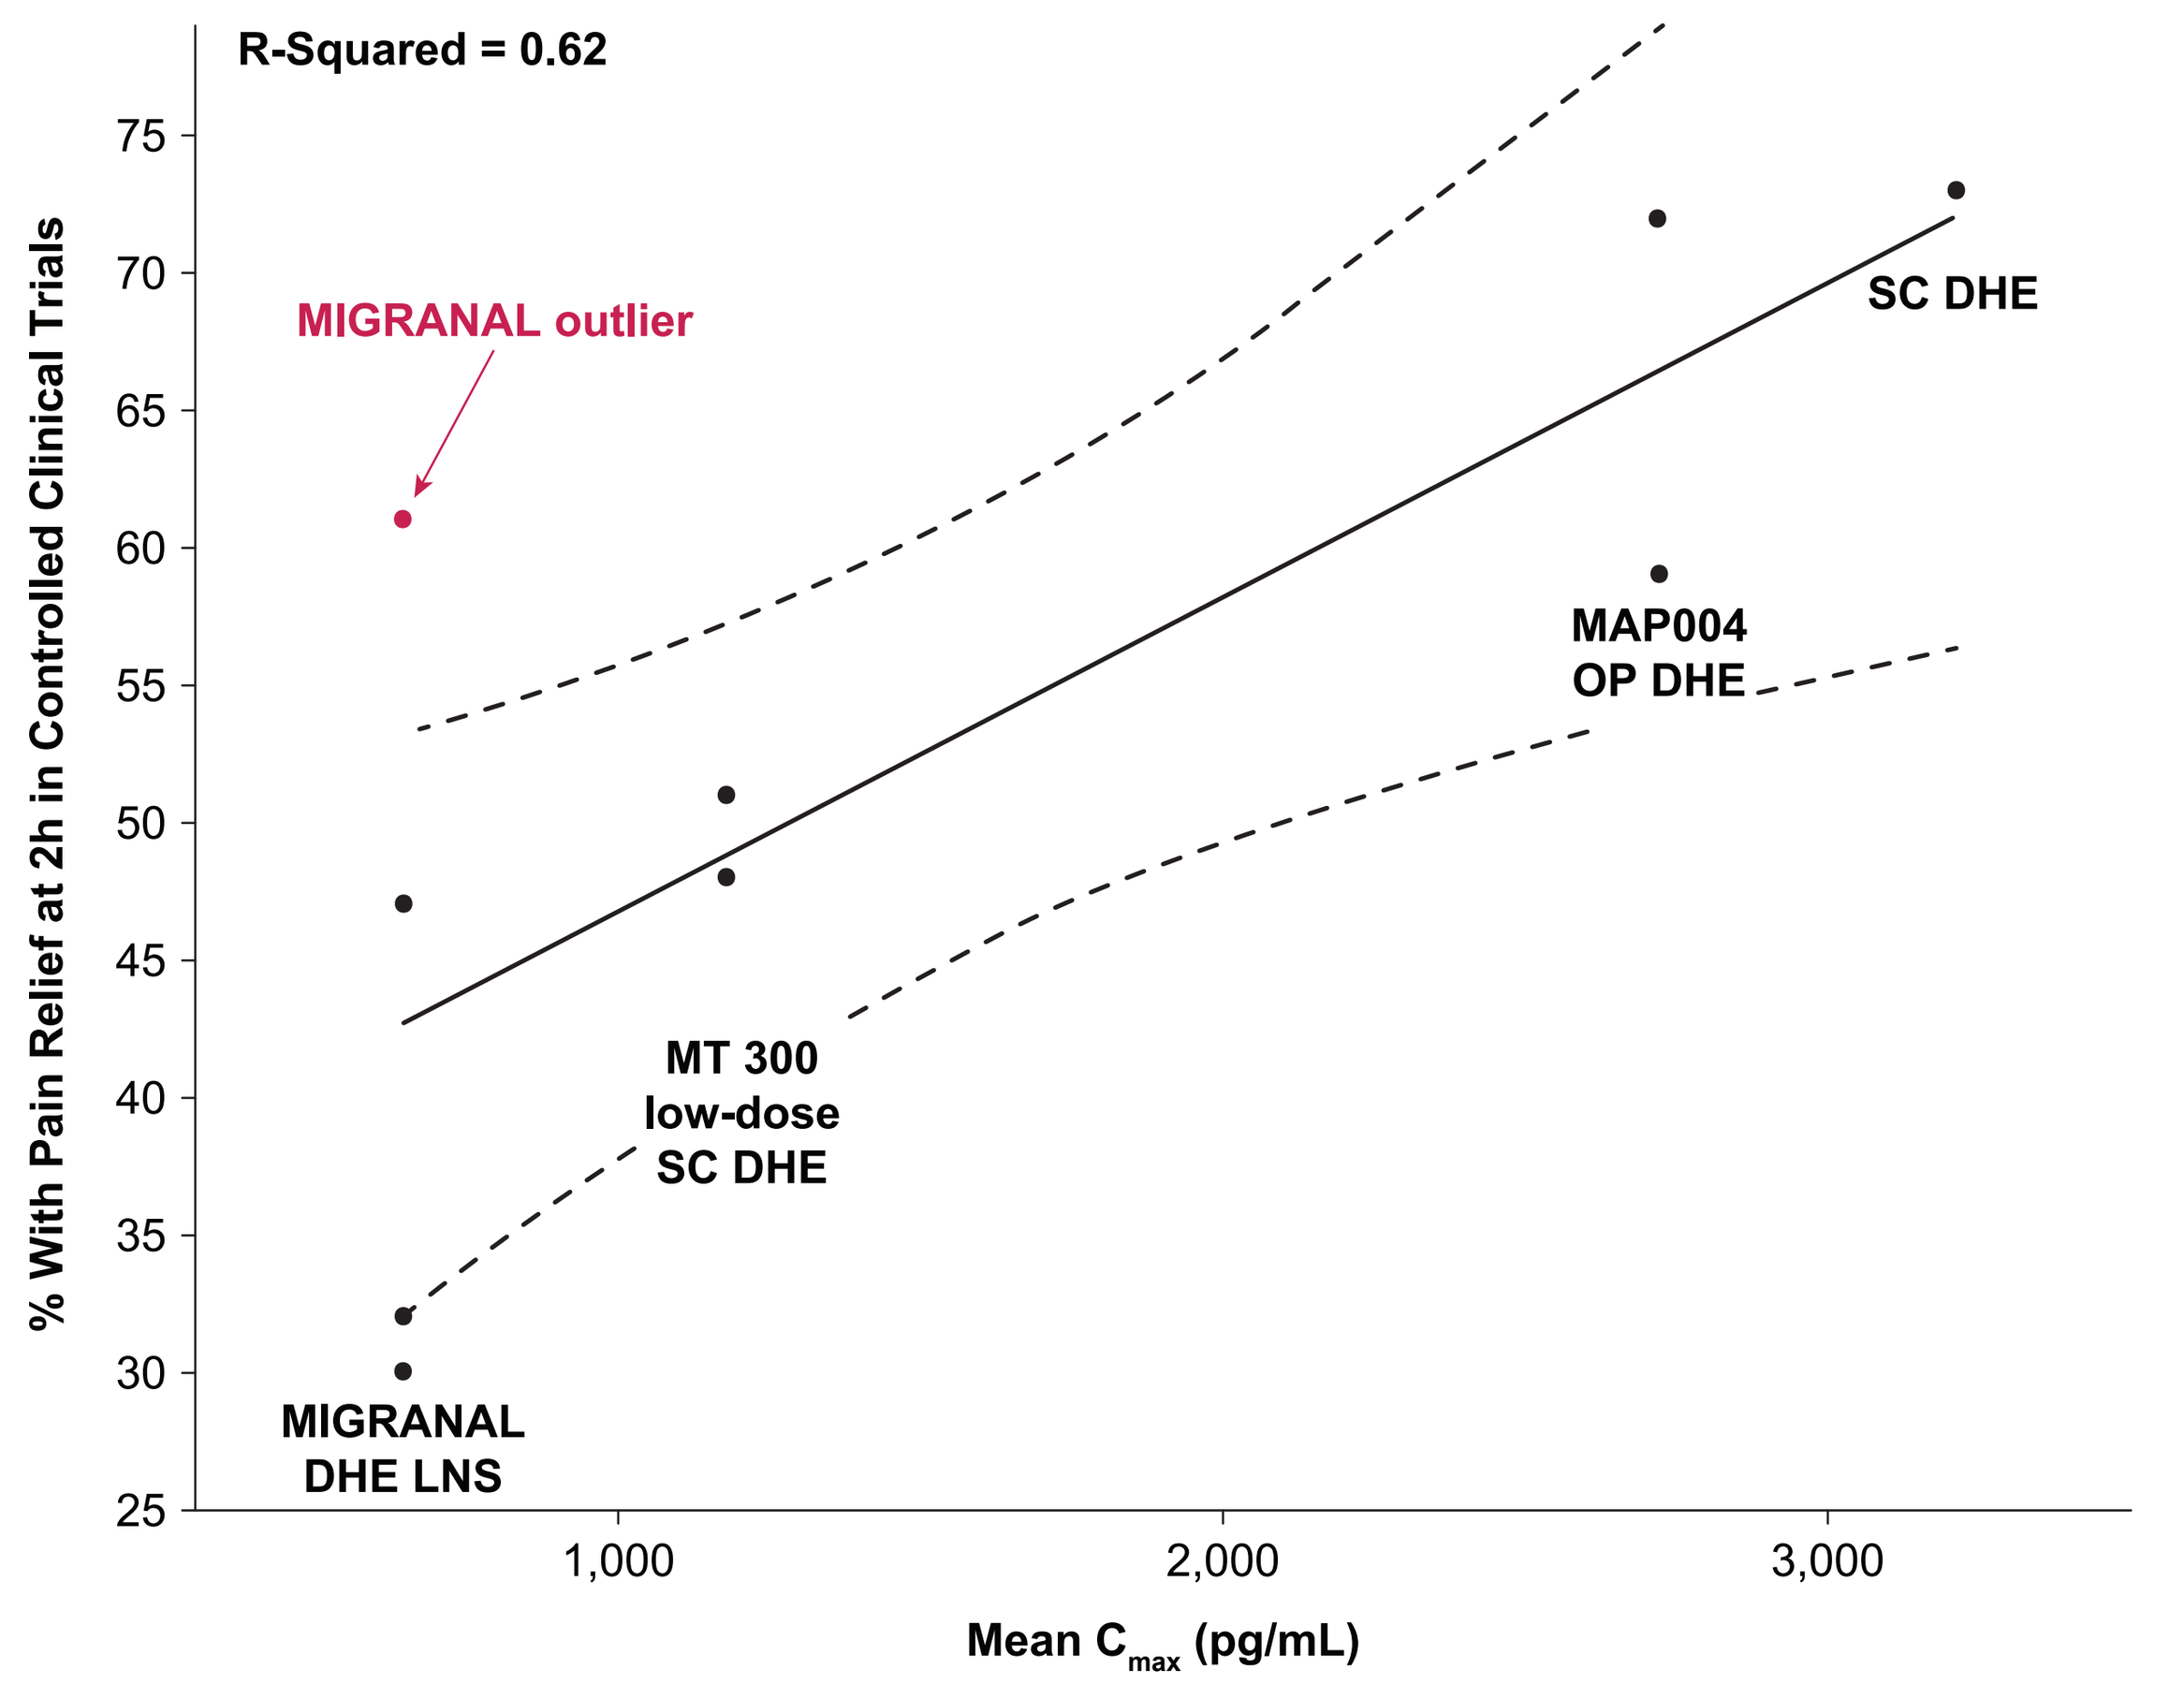
R-Squared = 0.62
% With Pain Relief at 2h in Controlled Clinical Trials
Mean C_{max} (pg/mL)
MIGRANAL outlier
SC DHE
MAP004 OP DHE
MT 300 low-dose SC DHE
MIGRANAL DHE LNS
25
30
35
40
45
50
55
60
65
70
75
1,000
2,000
3,000

Univariate linear regression analysis shows a relationship between the percentage of subjects with pain relief at 2 hours (clinical efficacy) compared to DHE C_{max} across 9 RCTs of various DHE formulations, doses, and routes of administration. The dotted lines represent the 95% confidence interval boundaries.
DHE, dihydroergotamine; LNS, liquid nasal spray; OP, oral pulmonary; SC, subcutaneous.

Figure 3. Relationship Between DHE $AUC_{0\text{-}0.5h}$ and 2-Hour Pain Relief

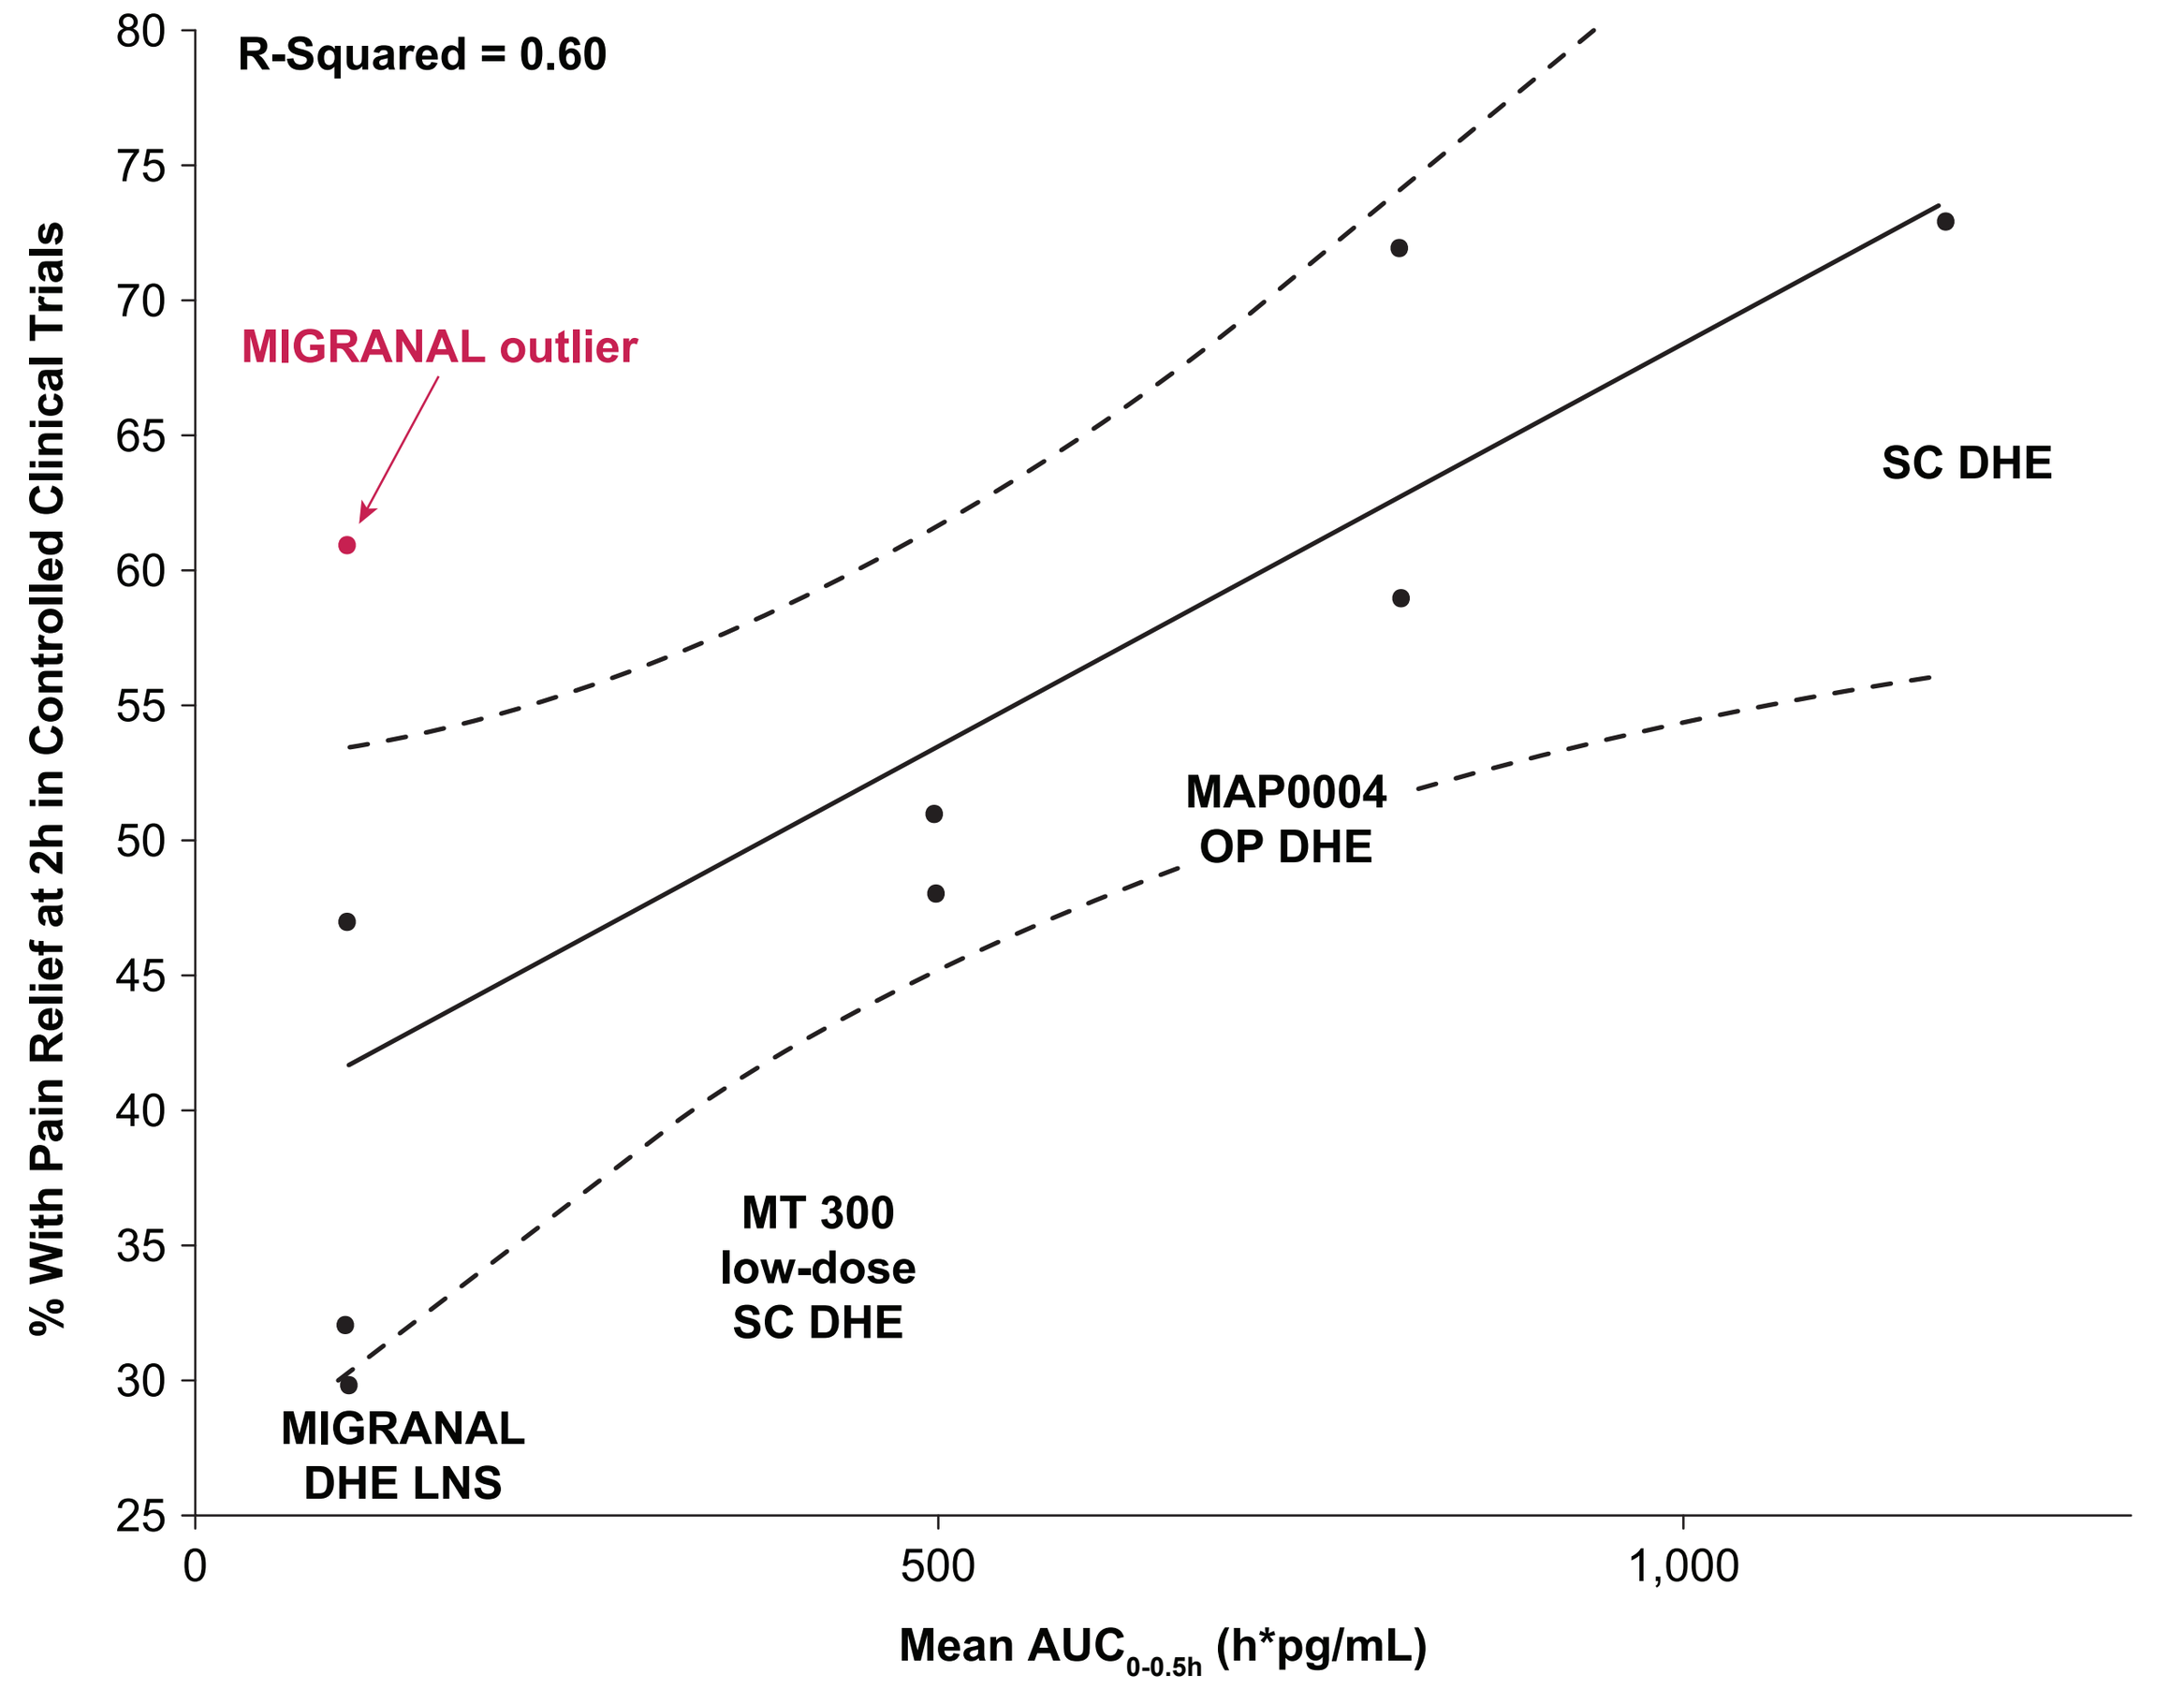
R-Squared = 0.60
% With Pain Relief at 2h in Controlled Clinical Trials
Mean $AUC_{0\text{-}0.5h}$ (h*pg/mL)
MIGRANAL outlier
SC DHE
MAP0004 OP DHE
MT 300 low-dose SC DHE
MIGRANAL DHE LNS
25
30
35
40
45
50
55
60
65
70
75
80
0
500
1,000

Univariate linear regression analysis shows a relationship between the percentage of subjects with pain relief at 2 hours (clinical efficacy) compared to DHE mean $AUC_{0\text{-}0.5h}$ across 9 RCTs of various DHE formulations, doses, and routes of administration. The dotted lines represent the 95% confidence interval boundaries.
DHE, dihydroergotamine; LNS, liquid nasal spray; OP, oral pulmonary; SC, subcuteneous.

Table 4. Relationship Between Mean DHE C_{max} and Nausea Incidence in a Phase 1 Study

Characteristic	LNS DHE (n=34)	STS101 5.2 mg (n=35)	STS101 7.0 mg (n=35)	STS101 8.6 mg (n=35)	IM DHE (n=34)
C_{max} (pg/mL) (SD)	673 (587)	2230 (823)	2710 (955)	2660 (1100)	3730 (801)
Nausea, n (%)	0	1 (2.9%)	3 (8.6%)	3 (8.6%)	3 (8.8%)

Within a single Phase 1 study, increasing mean DHE C_{max} was associated with increasing incidence of nausea.
DHE, dihydroergotamine; IM, intramuscular; LNS, liquid nasal spray; SD, standard deviation.

Figure 4. Relationship Between DHE C_{max} and Nausea Incidence across Phase 1 Studies

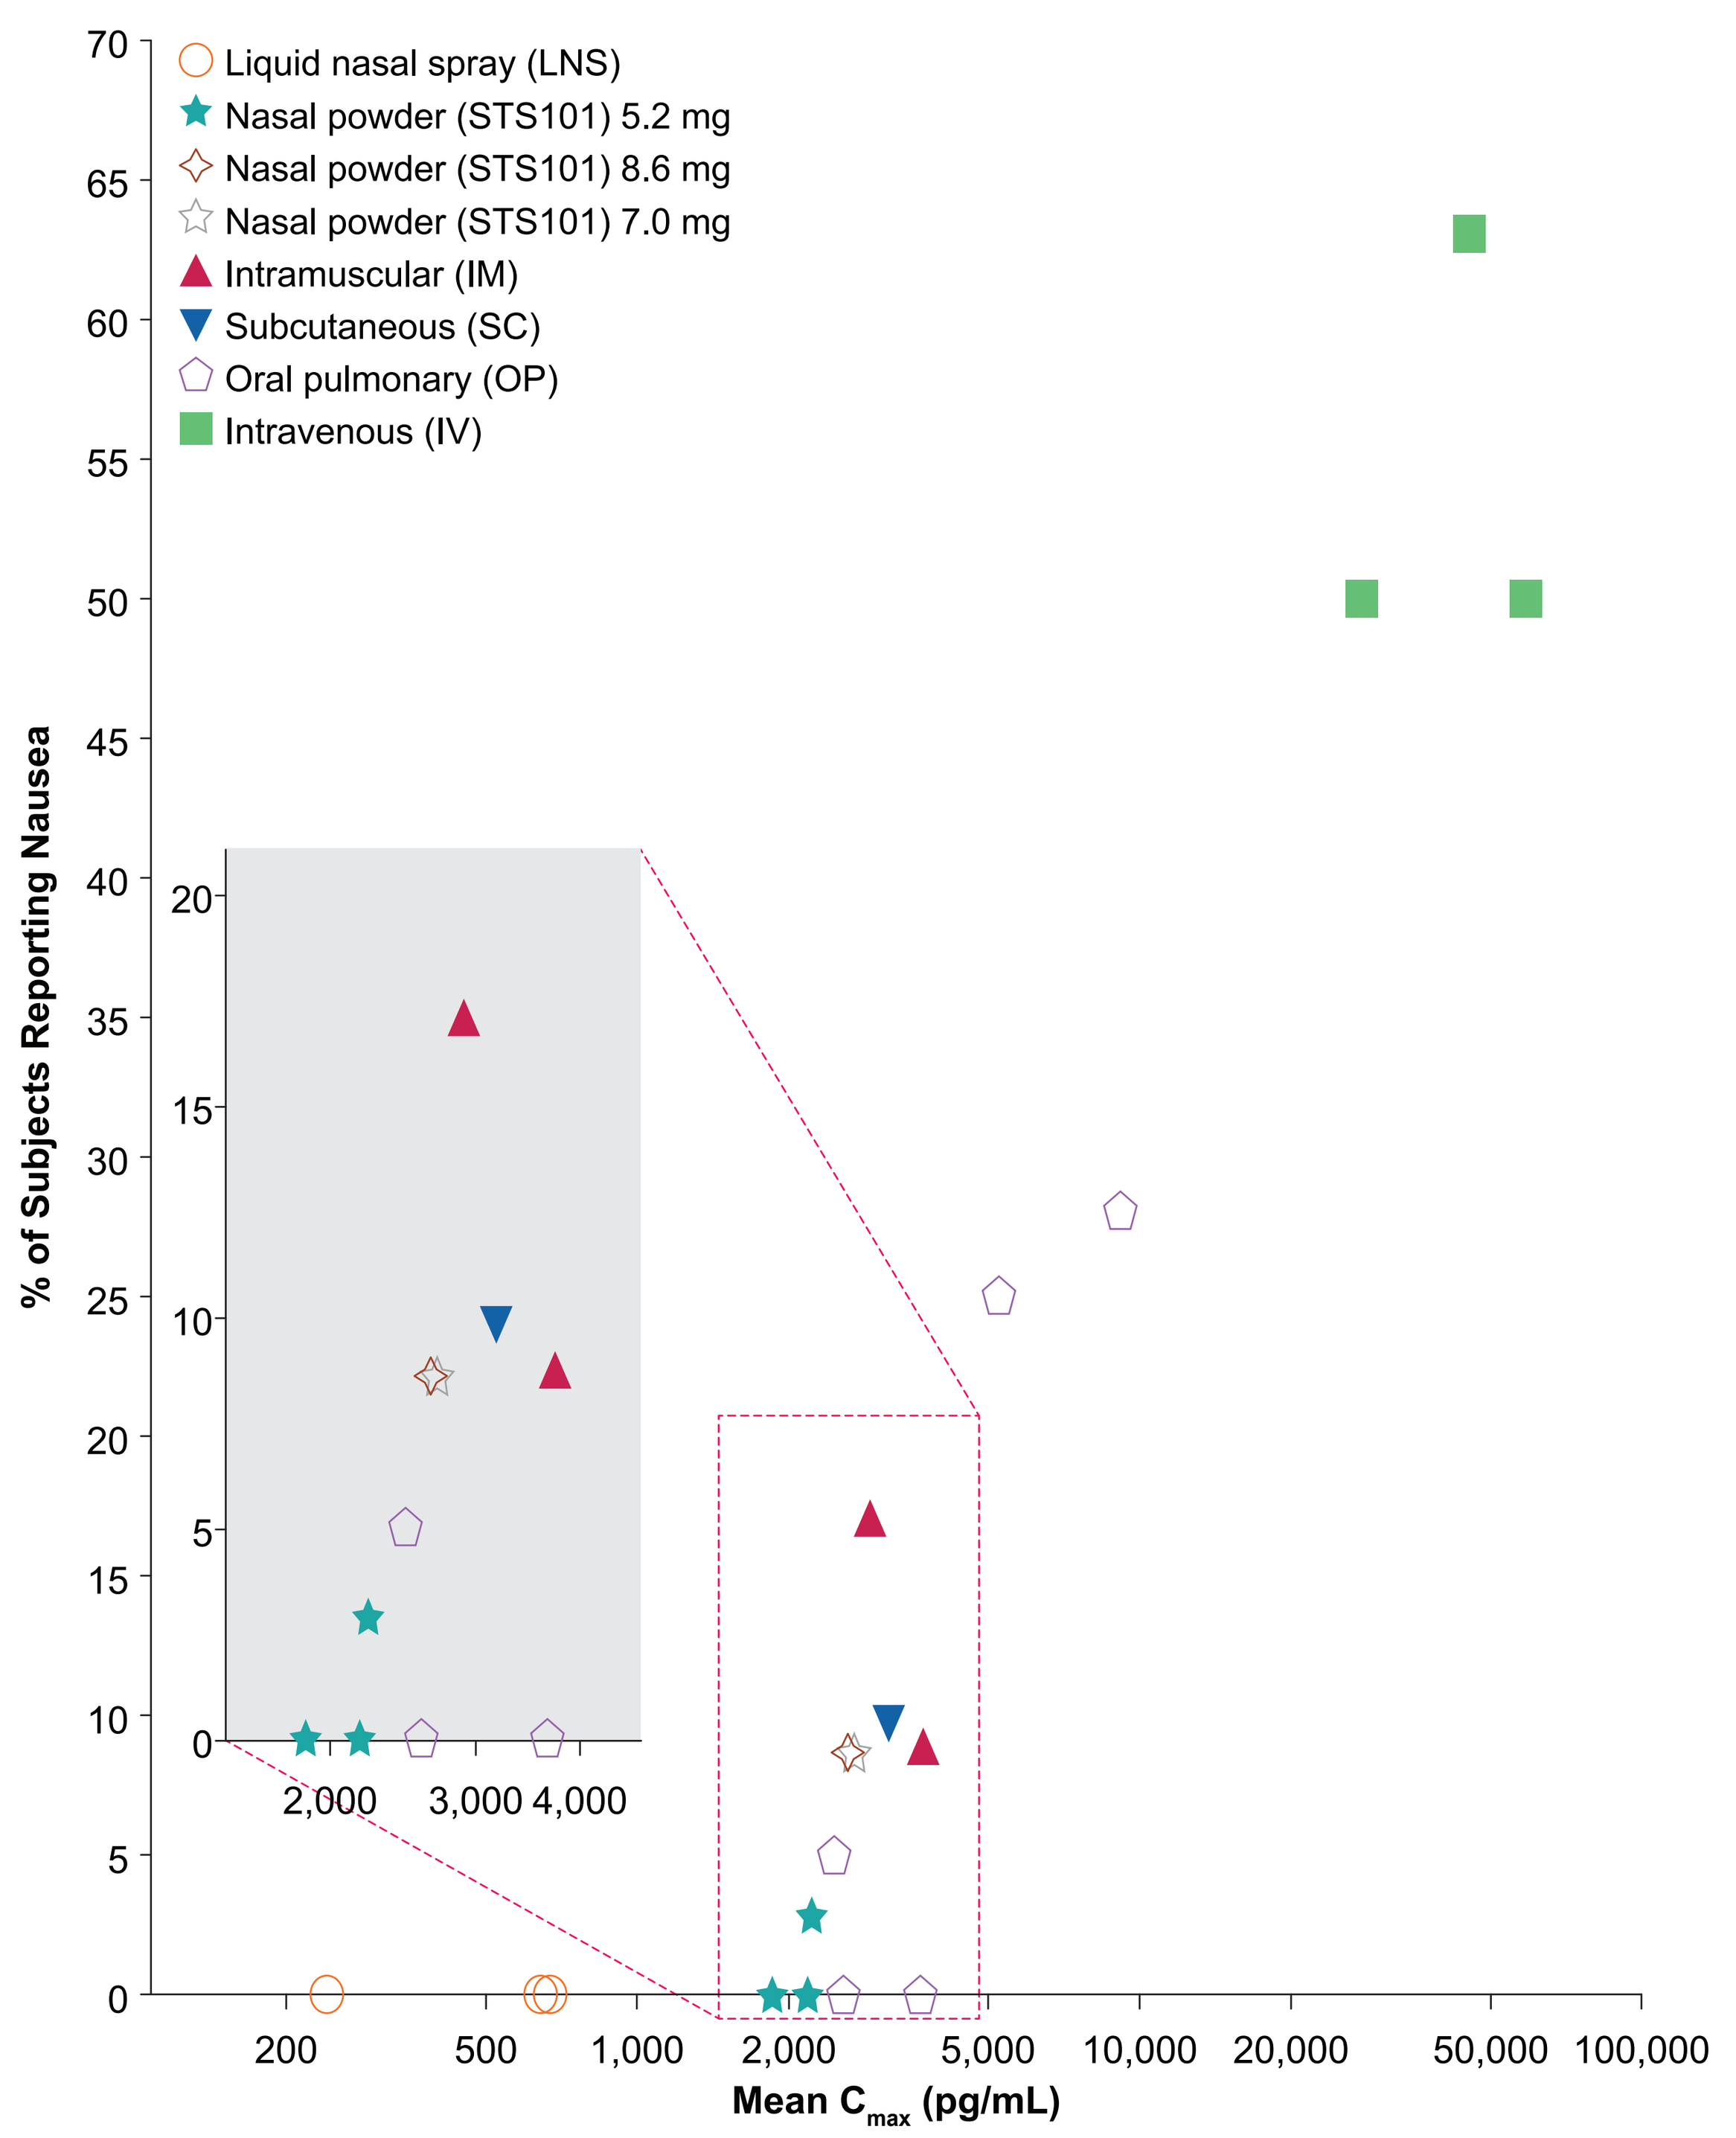
Liquid nasal spray (LNS)
Nasal powder (STS101) 5.2 mg
Nasal powder (STS101) 8.6 mg
Nasal powder (STS101) 7.0 mg
Intramuscular (IM)
Subcutaneous (SC)
Oral pulmonary (OP)
Intravenous (IV)
% of Subjects Reporting Nausea
Mean C_{max} (pg/mL)
0
5
10
15
20
25
30
35
40
45
50
55
60
65
70
200
500
1,000
2,000
5,000
10,000
20,000
50,000
100,000
2,000
3,000
4,000

The grey square represents the zoomed in area between ~2000 and 4500 pg/mL (as indicated by the dashed lines). In Phase 1 trials of DHE dosage forms with mean C_{max} values less than approximately 2500 pgml, nausea is infrequent or absent; above this threshold the incidence of nausea increases and is dependent upon mean C_{max}.

Conclusions

- DHE may have a PK "sweet spot": irrespective of route of administration or dosage form, DHE products that rapidly achieve, but do not exceed, a C_{max} in the range of approximately 2000 to 2500 pg/mL exhibit maximal clinical response rates at 2 hours attainable while keeping the incidence of the signature DHE side effect of nausea to a minimum.
- The PK profile of STS101 5.2 mg falls within this DHE "sweet spot," and the low incidence of nausea, vomiting, and other systemic side-effects reported in the ongoing STS101 open-label, long-term safety trial (Study STS101-003; ASCEND)[13] is consistent with the predictions from the analyses of Phase 1 trials detailed above.
 – Efficacy results from the ongoing Study STS101-007; SUMMIT will further elucidate relationships predicted here among DHE PK, efficacy and tolerability

References

1. Silberstein SD, et al. *Headache.* 2020;60(1):40-57.
2. Horton BT, et al. *Proc Staff Meet Mayo Clin.* 1945;20:241-248.
3. Silberstein SD, et al. *Headache.* 2003;42(3):144-66.
4. Marmura MJ, et al. *Headache.* 2015;55(1):3-20.
5. Novartis. DHE 45 US Prescribing Information. 2017.
6. Bausch Health. Migranal Prescribing Information. 2019.
7. Impel NeuroPharma. Trudhesa Prescribing Information. 2021.
8. Kellerman D, et al. *Neurology.* 2012;78:P03.238
9. Albrecht D, et al. AAN 2022 Annual Meeting Abstracts. 2022:P13.003.
10. Gabrielsson J, et al. *Methods Mol Bio.* 2012:377-389.
11. Gallagher RM, et al. *Arch Neuro.* 1996;53(12):1285-91.
12. Tepper SJ, et al. AAN 2022 Annual Meeting Abstracts. 2022:IOR-08.

Disclosures

John Kollins, Shannon Strom, and *Detlef Albrecht* are employees and/or stockholders of Satsuma Pharmaceuticals, Inc.

Acknowledgements

Funding for this study and poster support was provided by Satsuma Pharmaceuticals, Inc. (South San Francisco, CA, USA). Medical writing and editorial support were provided by The Medicine Group, LLC (New Hope, PA, USA) and in accordance with Good Publication Practice guidelines.

Presented at the American Headache Society (AHS) Annual Meeting| June 9–12, 2022 | Denver, CO.