P-612

STS101 Demonstrated Long-Term Clinical Benefit in the Phase 3 Open-Label ASCEND Study

Larry Charleston IV, MD, MSc, FAHS[1], Shannon Strom, PhD[2], Detlef Albrecht, MD[2], Robert P. Cowan, MD[3]

[1]Michigan State University College of Human Medicine, Grand Rapids, MI, USA; [2]Satsuma Pharmaceuticals, Inc., Durham, NC, USA; [3]Stanford University, Stanford, CA, USA

Introduction

- Dihydroergotamine mesylate (DHE) is a recommended first-line treatment option for the acute treatment of moderate or severe migraine attacks, with or without aura.[1]
- STS101 is a novel investigational DHE product that combines a mucoadhesive nasal powder formulation delivered with an easy-to-use, easy-to-carry, pre-filled, single-use nasal delivery device (Figure 1).
- The STS101 advanced nasal powder and device technology maximizes deposition of DHE on the nasal mucosa, enhancing DHE absorption, increasing drug exposure and reducing pharmacokinetic variability in comparison with DHE liquid nasal sprays.
- The ASCEND study primarily assessed the safety and tolerability of STS101 5.2 mg in the acute treatment of migraine attacks with or without aura over 12 months. The secondary objective was to describe the effectiveness of STS101 over 12 months.

Objective

- To describe the long-term effectiveness of STS101 over 12 months in the open-label ASCEND study

Methods

Study Design and Treatment Intervention

- ASCEND was a multi-center, multi-dose, open-label, 12-month study of STS101 in adults aged 18–65 years with migraine (NCT04406649).
- After establishing eligibility, the participants could self-administer STS101 5.2 mg as needed (PRN) for up to 2 doses within 24 hours to treat a single migraine attack, and up to 12 doses/month for 12 months.
- As an open-label study, all effectiveness analyses were exploratory.

Participants

- Study participants must have had ≥1-year history of migraine (with or without aura) according to the International Classification of Headache Disorders, 3rd edition,[2] including:
 - Migraine onset before age of 50 years
 - 4–12 migraine attacks/month in each of the 3 months prior to screening
 - <15 headache days/month in each of the 3 months prior to screening
- Exclusion criteria included diagnosis of non-migraine headache, history of cerebrovascular disease, and ≥2 cardiovascular risk factors.
- Participants must have had an intact nasal mucosa at baseline (i.e., no ulceration or bleeding; no or mild erythema, swelling, or rhinorrhea).
- Effectiveness data were analyzed in either the expansion population or modified intent-to-treat (mITT) population.
 - Expansion population: All participants who enrolled in the expansion phase of the study and exclusively used the STS101 delivery device submitted for FDA approval
 - mITT population: All participants who treated ≥1 migraine attack with study medication from the final version of STS101 and had ≥1 post-baseline effectiveness value

Effectiveness Outcomes

- Proportion of migraine attacks with headache pain freedom
 - Defined as a reduction from a baseline pain severity score of 2 (moderate) or 3 (severe) on a 4-point scale to a score of 0 (none)
 - Required a non-missing response at the analyzed time point and no use of rescue medication or a second dose of study medication prior to the analyzed time point
 - Analyzed in the expansion population
- Proportion of migraine attacks free from most bothersome symptom (MBS)
 - Defined as the absence of the baseline MBS (photophobia, phonophobia, or nausea)
 - Required a non-missing response at the analyzed time point and no use of rescue medication or a second dose of study medication prior to the analyzed time point
 - Analyzed in the expansion population
- Proportion of migraine attacks with headache pain relief
 - Defined as a reduction from a baseline pain severity score of 2 (moderate) or 3 (severe) on a 4-point scale to a score of 0 (none) or 1 (mild)
 - Required a non-missing response at the analyzed time point and no use of rescue medication or a second dose of study medication prior to the analyzed time point
 - Analyzed in the mITT population
- Proportion of migraine attacks with rescue medication use
 - Defined as any medication taken to treat migraine after dosing with study medication
 - Analyzed in the mITT population

Results

- 446 participants were enrolled and treated 9,091 migraine attacks over 12 months.
 - 172 participants exclusively used the nasal delivery device submitted for FDA approval to treat 3,394 migraine attacks and were included in the expansion population (Table 1).
- The proportion of migraine attacks with response increased through 24 hours and was stable at 48 hours post-dose for freedom from headache pain (Figure 2), freedom from MBS (Figure 3), and relief from headache pain (Figure 4).
- The proportion of migraine attacks free from headache pain at 2 hours (Figure 5), free from MBS at 2 hours (Figure 6), and with relief from headache pain at 2 hours (Figure 7) were stable throughout the 12-month treatment period.
- Rescue medication was used in 3.5% and 4.6% of migraine attacks within 24 and 48 hours post-dose, respectively.
- A second dose of study medication was used in 19.3% and 19.4% of migraine attacks within 24 and 48 hours post-dose, respectively.

Figure 1. STS101 Administration

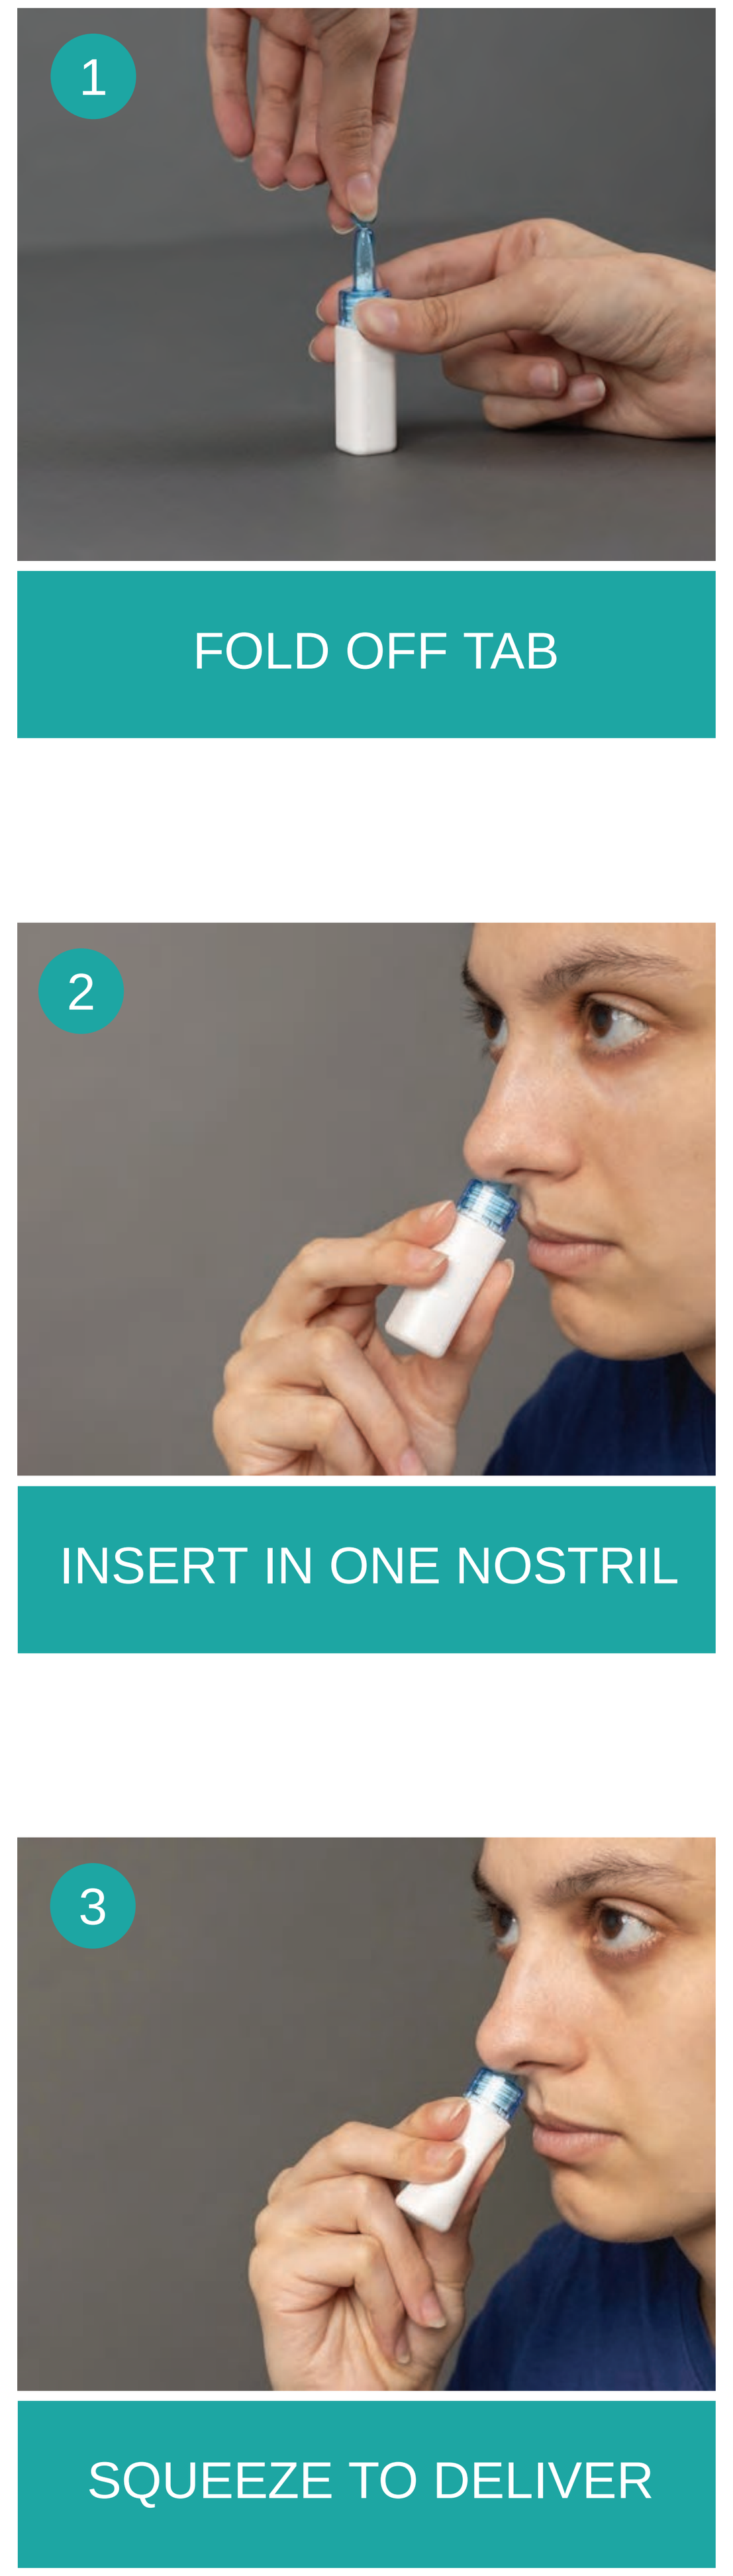
1
FOLD OFF TAB
2
INSERT IN ONE NOSTRIL
3
SQUEEZE TO DELIVER

Table 1. Baseline Demographics and Migraine Characteristics

	Expansion Population Patients, N=172
Mean (SD) age, y	39.1 (11.0)
Sex, n (%)	
Female	144 (83.7)
Male	28 (16.3)
Ethnicity, n (%)	
Hispanic or Latino	74 (43.0)
Not Hispanic or Latino	98 (57.0)
Race, n (%)	
White	150 (87.2)
Black or African American	18 (10.5)
Asian	3 (1.7)
Other	1 (0.6)
Mean (SD) weight, kg	75.5 (16.0)
Mean (SD) height, cm	165.8 (9.1)
Mean (SD) BMI, kg/m²	27.3 (5.0)
	Attacks, N=3389
Baseline migraine severity, n (%)	
Mild	204 (6.0)
Moderate	1607 (47.4)
Severe	1578 (46.6)
Nausea at baseline, n (%)	2235 (65.9)
Photophobia at baseline, n (%)	3269 (96.5)
Phonophobia at baseline, n (%)	3197 (94.3)

BMI, body mass index; SD, standard deviation.

Figure 2. Proportion of Migraine Attacks With Headache Pain Freedom Throughout 48 Hours Post-Dose

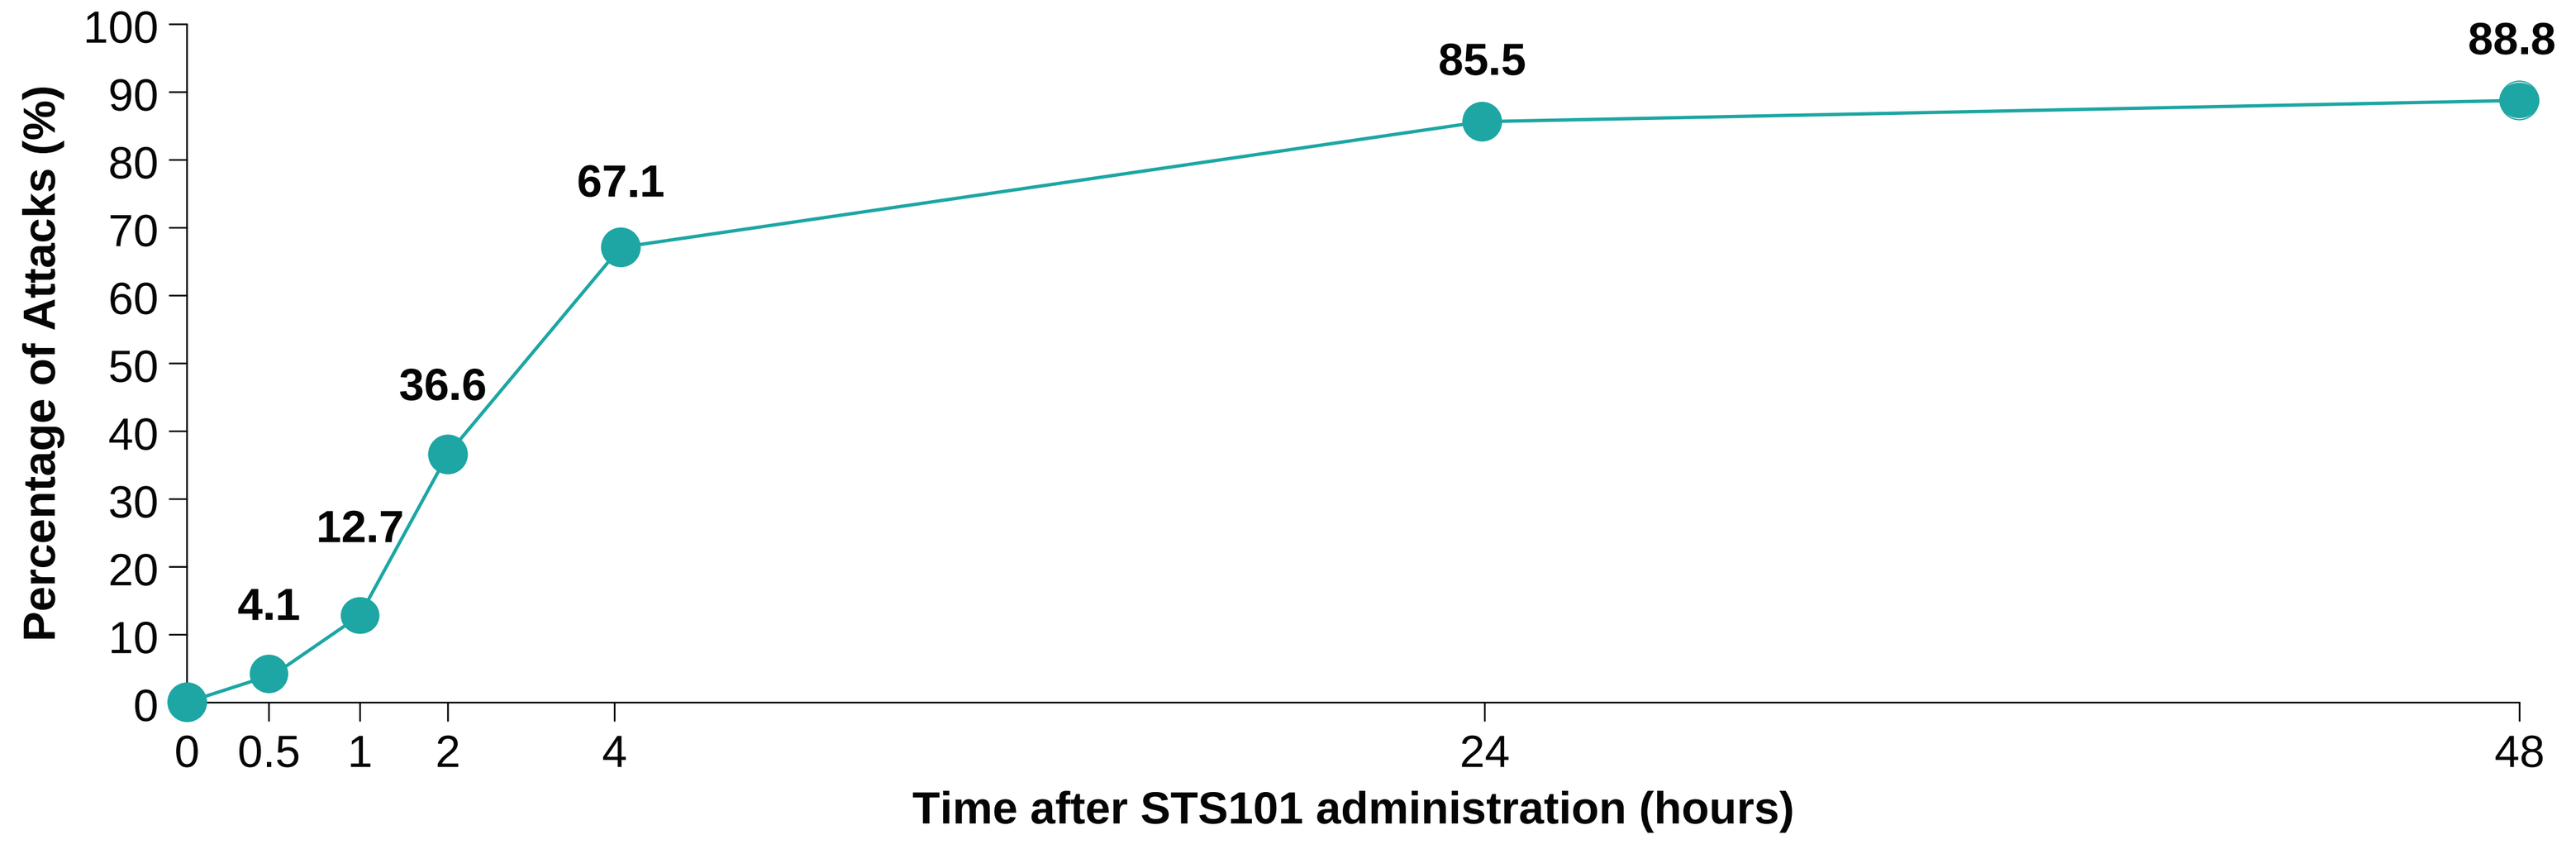
Percentage of Attacks (%)
0
10
20
30
40
50
60
70
80
90
100
4.1
12.7
36.6
67.1
85.5
88.8
0
0.5
1
2
4
24
48
Time after STS101 administration (hours)

Analyzed in the expansion population (all participants who enrolled in the expansion phase of the study and exclusively used the final version of STS101).

Figure 3. Proportion of Migraine Attacks Free From MBS Throughout 48 Hours Post-Dose

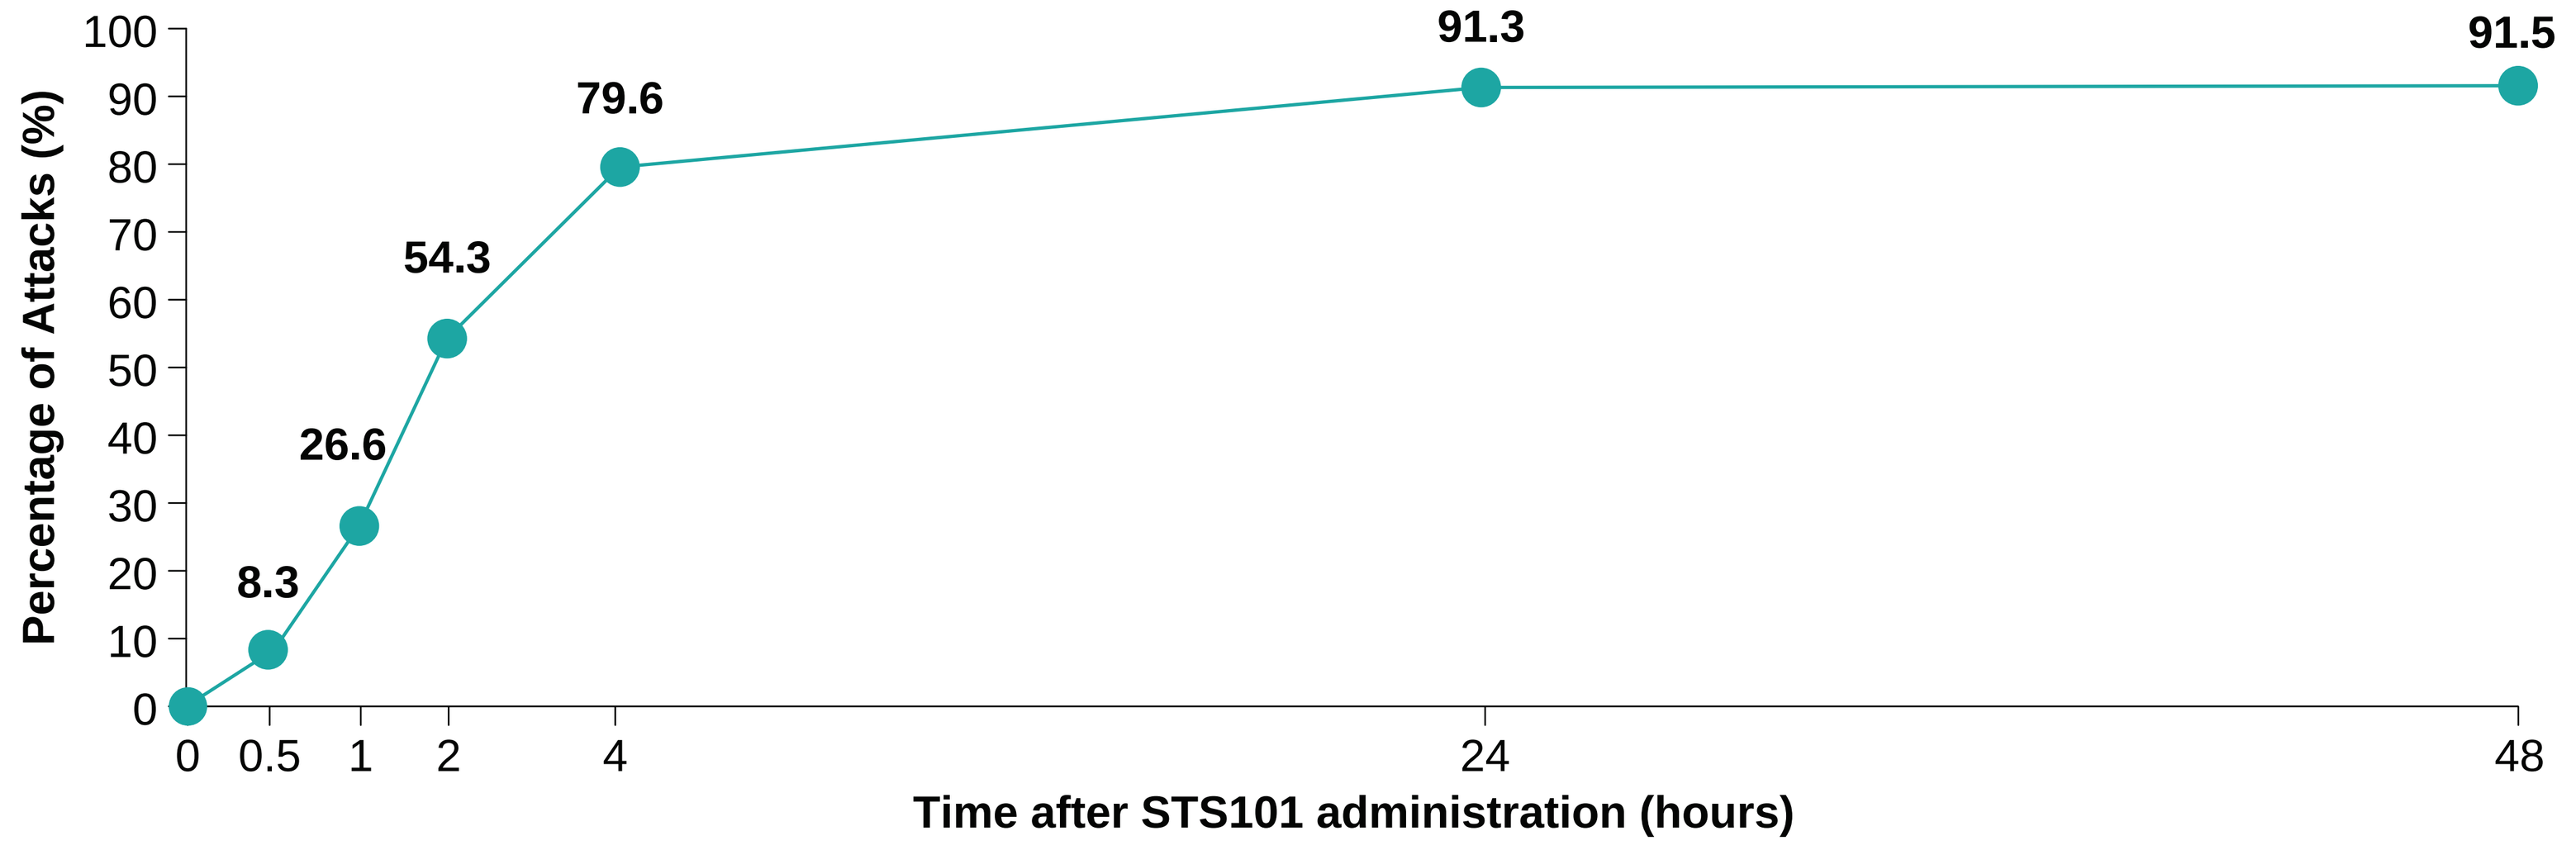
Percentage of Attacks (%)
0
10
20
30
40
50
60
70
80
90
100
8.3
26.6
54.3
79.6
91.3
91.5
0
0.5
1
2
4
24
48
Time after STS101 administration (hours)

Analyzed in the expansion population (all participants who enrolled in the expansion phase of the study and exclusively used the final version of STS101). MBS, most bothersome symptom.

Figure 4. Proportion of Migraine Attacks With Headache Pain Relief Throughout 48 Hours Post-Dose

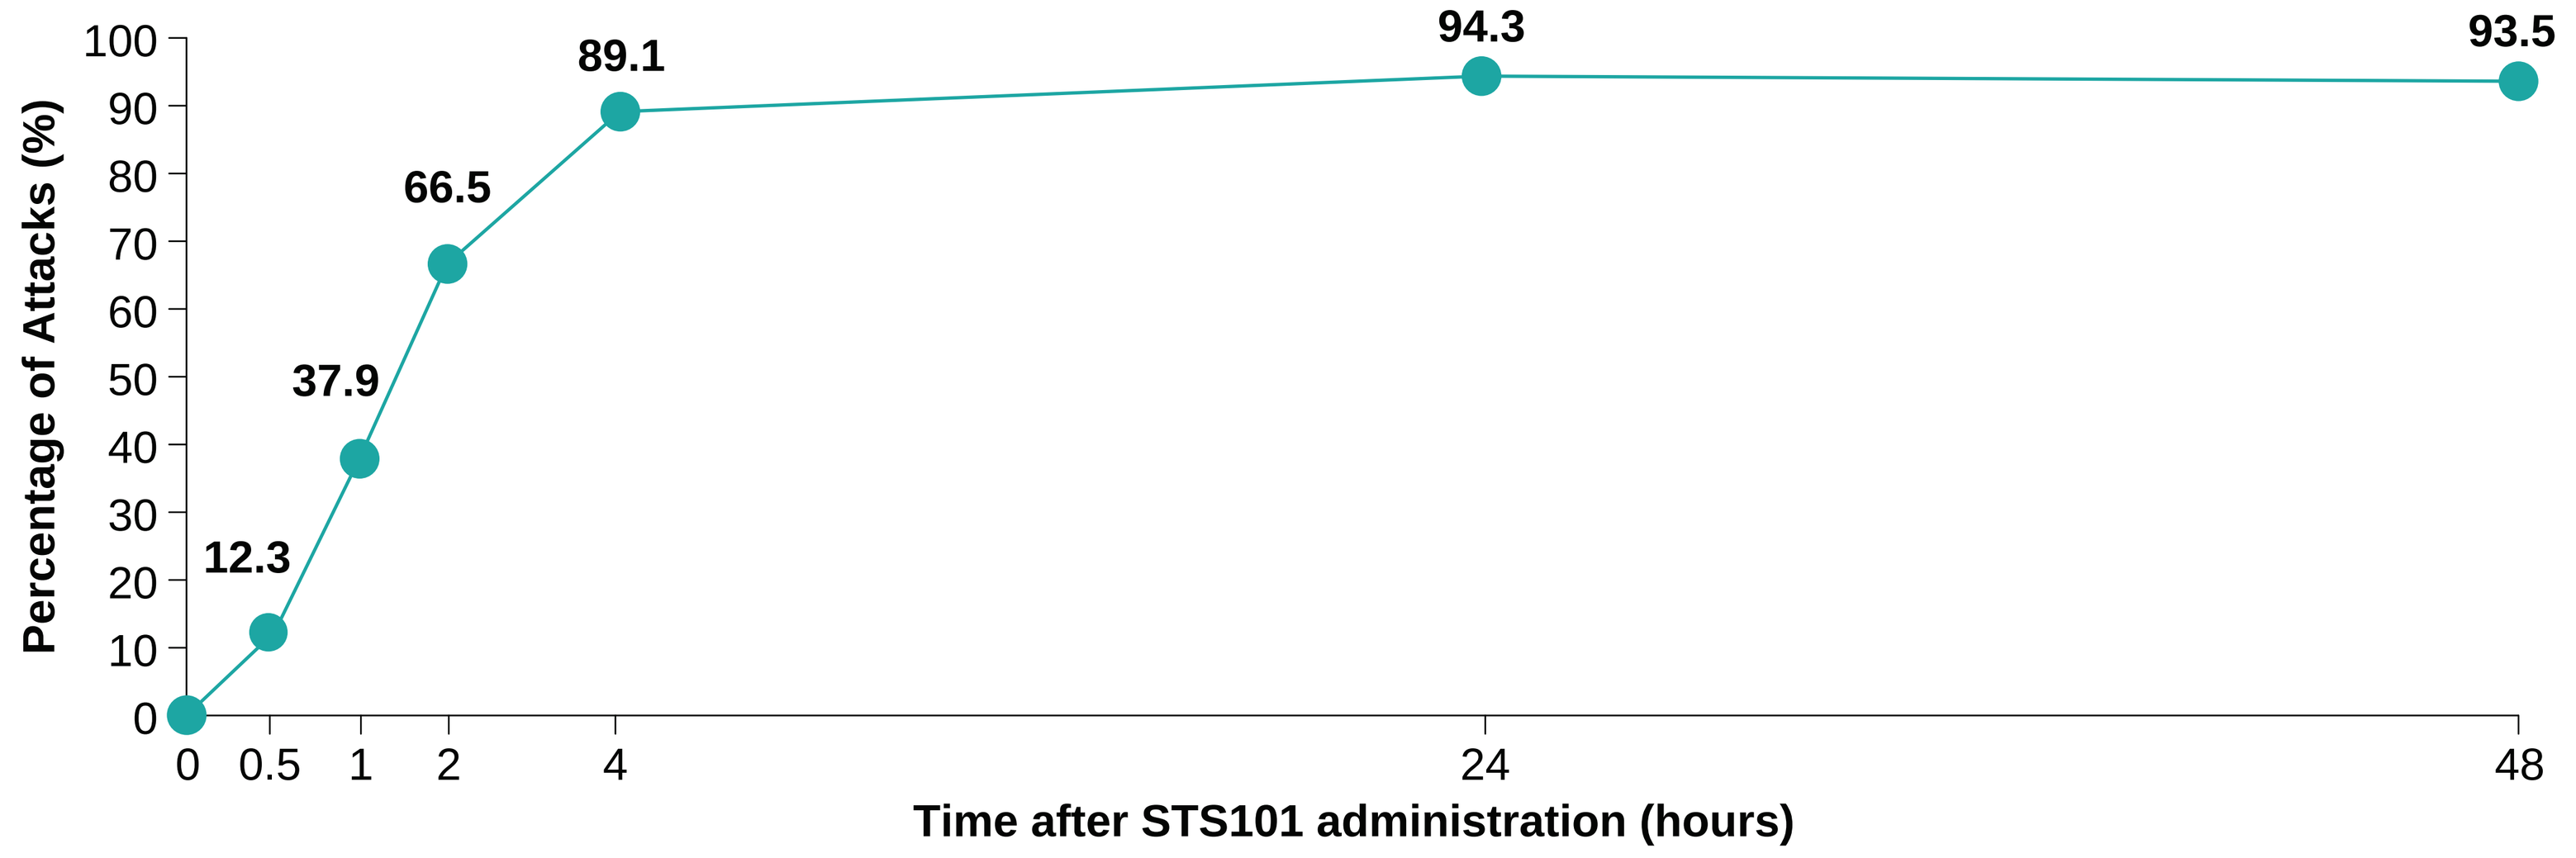
Percentage of Attacks (%)
0
10
20
30
40
50
60
70
80
90
100
12.3
37.9
66.5
89.1
94.3
93.5
0
0.5
1
2
4
24
48
Time after STS101 administration (hours)

Analyzed in the expansion population (all participants who enrolled in the expansion phase of the study and exclusively used the final version of STS101).

Figure 5. Proportion of Migraine Attacks With Headache Pain Freedom at 2 Hours Post-Dose, by Month Over 12 Months

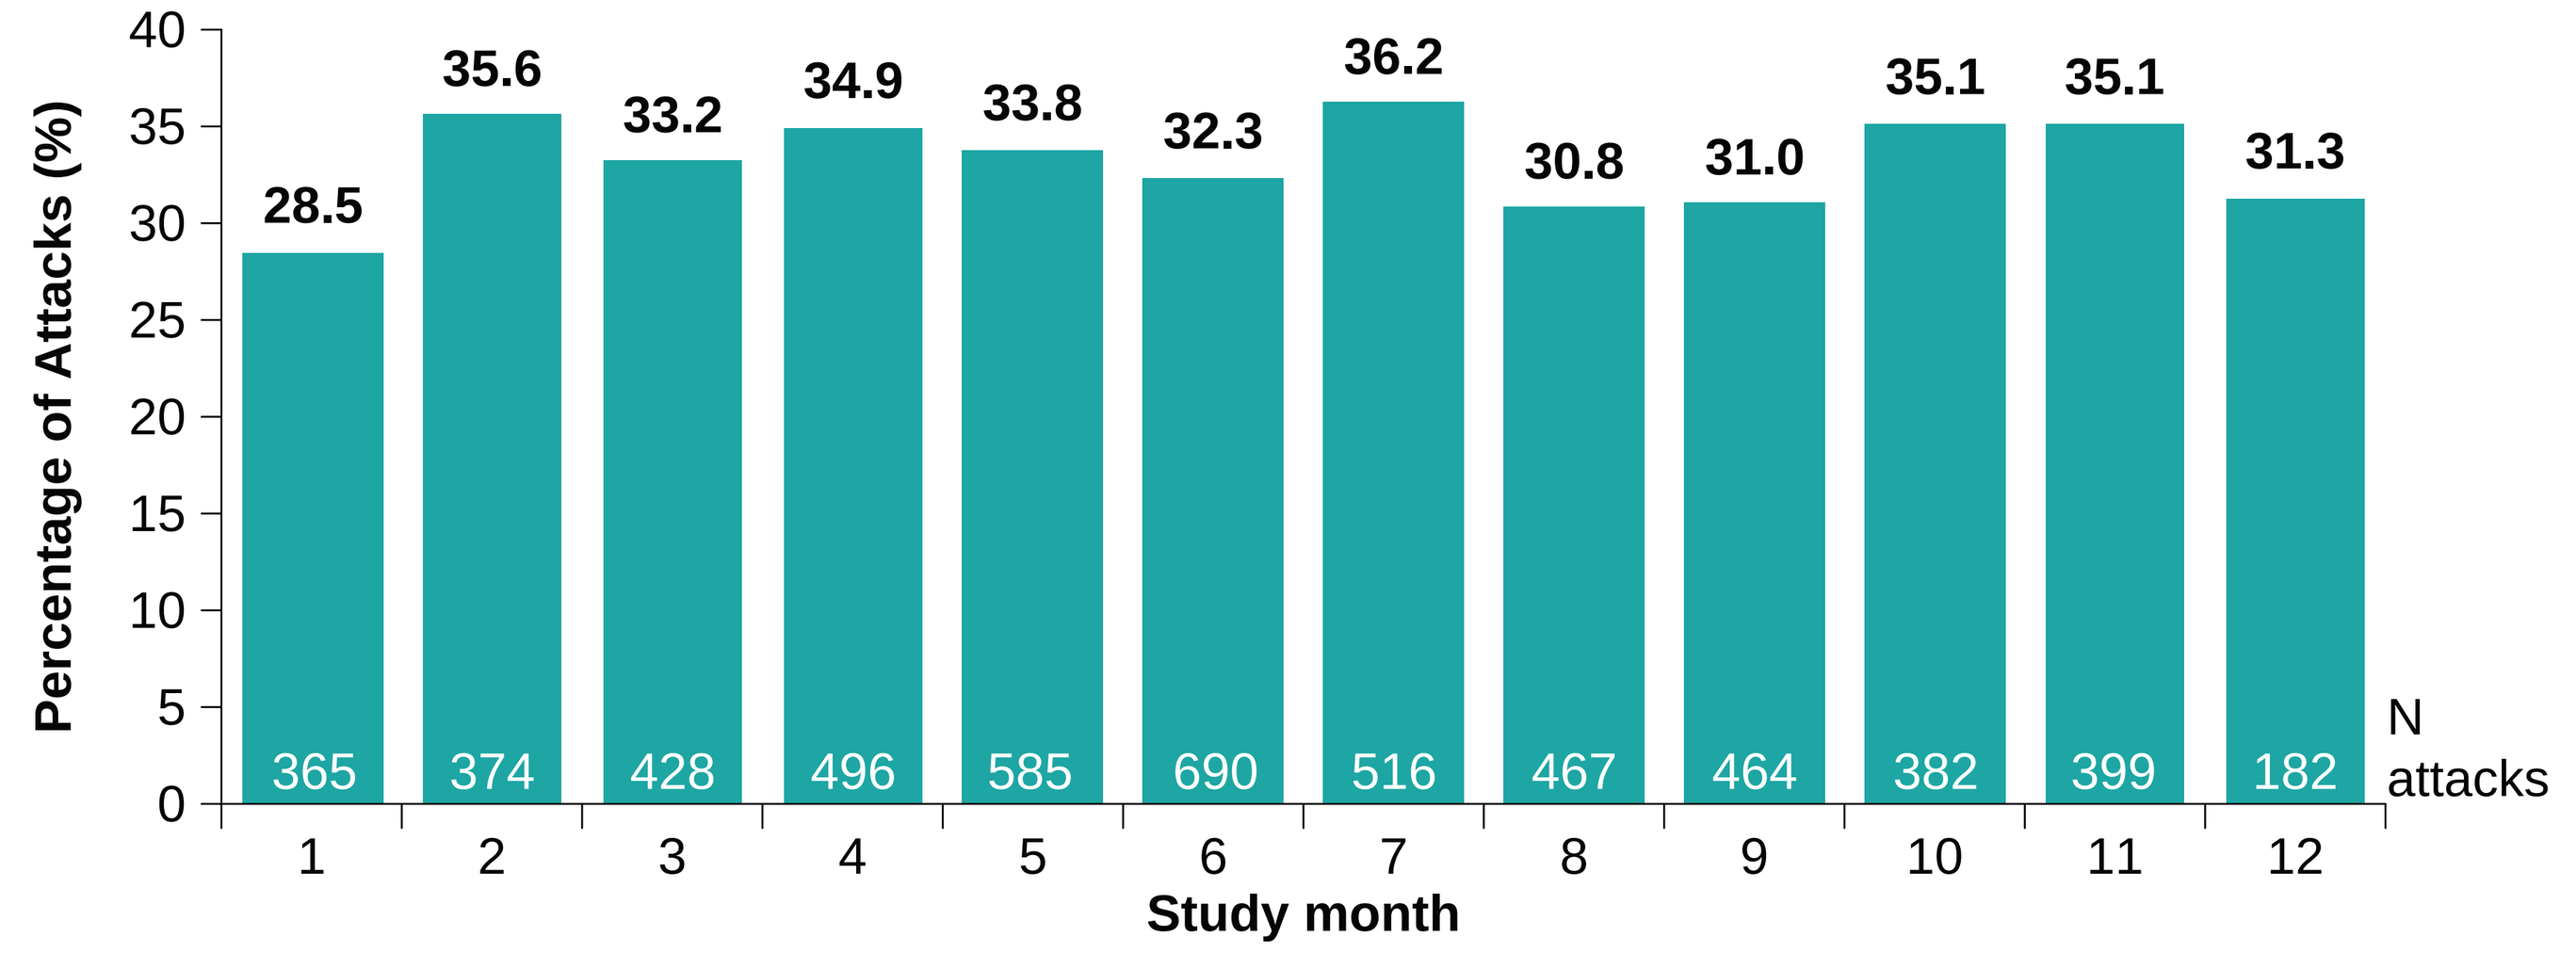
Percentage of Attacks (%)
0
5
10
15
20
25
30
35
40
28.5
35.6
33.2
34.9
33.8
32.3
36.2
30.8
31.0
35.1
35.1
31.3
365
374
428
496
585
690
516
467
464
382
399
182
N attacks
1
2
3
4
5
6
7
8
9
10
11
12
Study month

Analyzed in the modified intent-to-treat population that treated ≥1 migraine attack with study medication from the final version of STS101 and had ≥1 post-baseline effectiveness value at any time point.

Figure 6. Proportion of Migraine Attacks Free From MBS at 2 Hours Post-Dose, by Month Over 12 Months

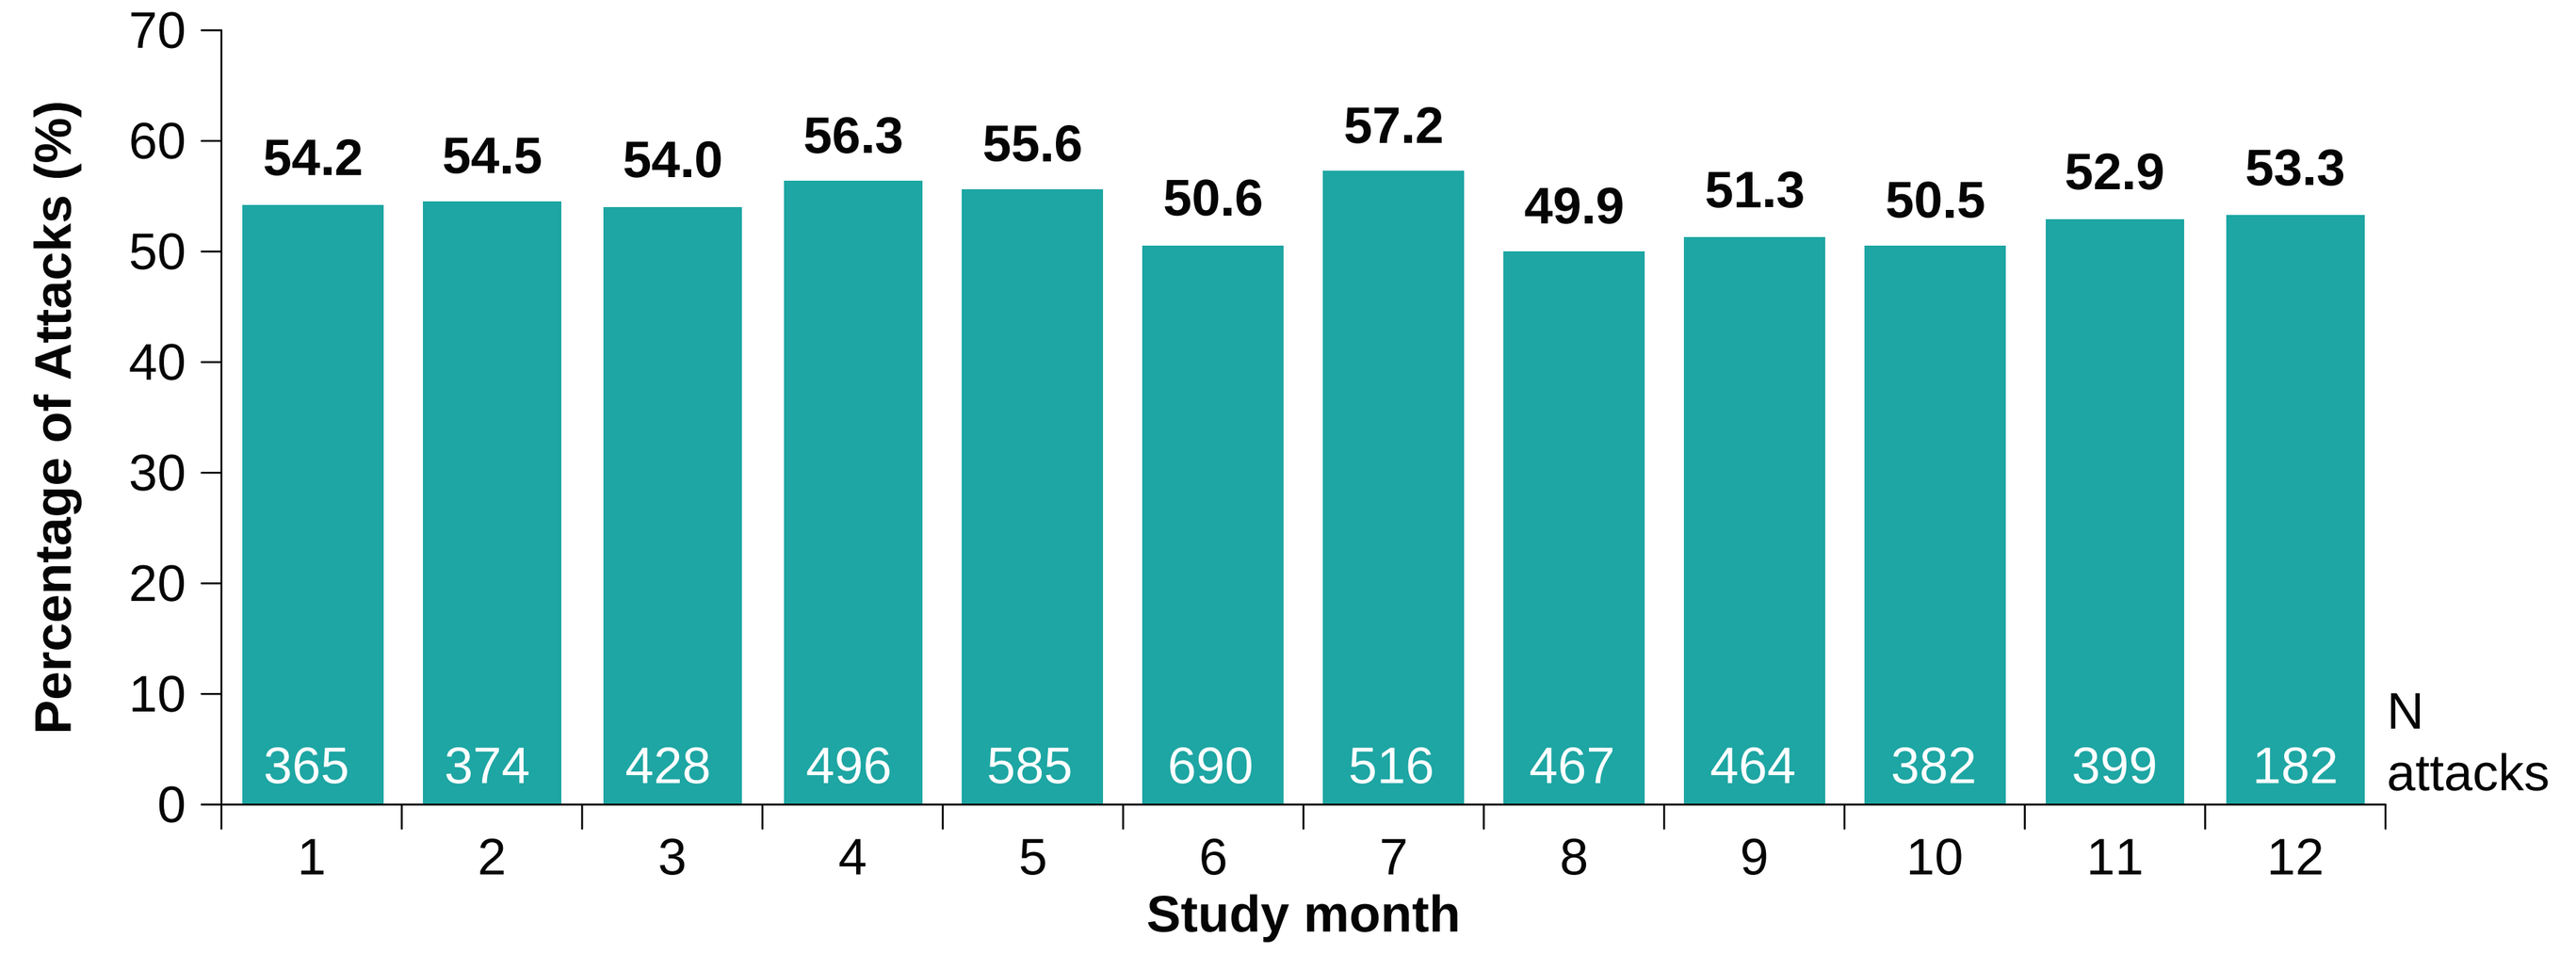
Percentage of Attacks (%)
0
10
20
30
40
50
60
70
54.2
54.5
54.0
56.3
55.6
50.6
57.2
49.9
51.3
50.5
52.9
53.3
365
374
428
496
585
690
516
467
464
382
399
182
N attacks
1
2
3
4
5
6
7
8
9
10
11
12
Study month

Analyzed in the modified intent-to-treat population that treated ≥1 migraine attack with study medication from the final version of STS101 and had ≥1 post-baseline effectiveness value at any time point.

Figure 7. Proportion of Migraine Attacks With Headache Pain Relief at 2 Hours Post-Dose, by Month Over 12 Months

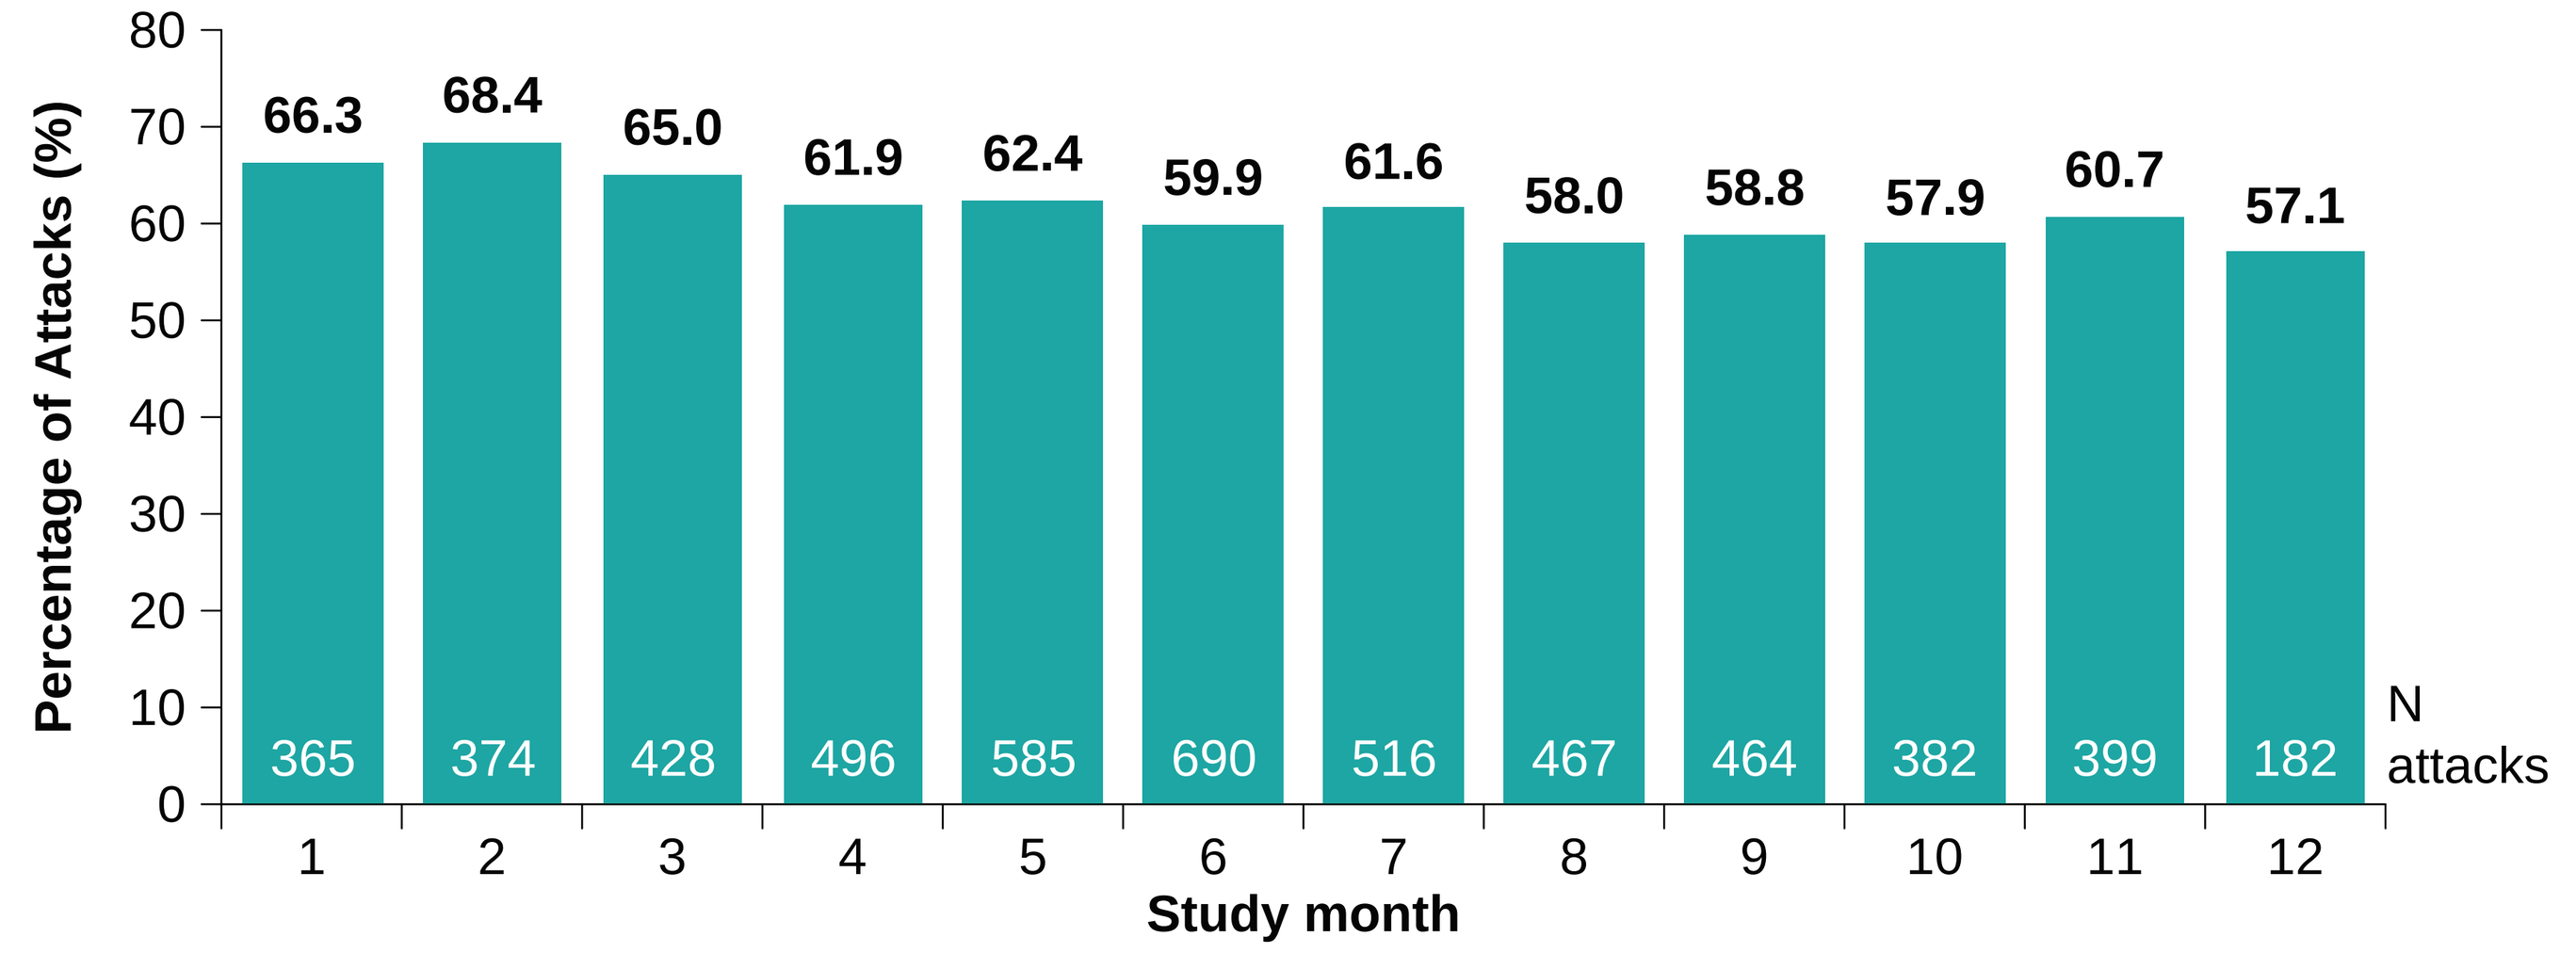
Percentage of Attacks (%)
0
10
20
30
40
50
60
70
80
66.3
68.4
65.0
61.9
62.4
59.9
61.6
58.0
58.8
57.9
60.7
57.1
365
374
428
496
585
690
516
467
464
382
399
182
N attacks
1
2
3
4
5
6
7
8
9
10
11
12
Study month

Analyzed in the modified intent-to-treat population that treated ≥1 migraine attack with study medication from the final version of STS101 and had ≥1 post-baseline effectiveness value at any time point.

Conclusions

- Exploratory effectiveness analyses in this long-term safety study showed high rates of freedom from headache pain and MBS through 48 hours post-dose for STS101.
- These effects were consistent throughout 12 months of study showing that STS101 can be a reliable and effective long-term treatment for acute migraine attacks.
- Headache pain relief at 2 hours post-dose occurred in up to two-thirds of all treated migraine attacks and was relatively stable throughout 12 months of study.

References

1. Ailani J, et al. *Headache.* 2021;61(7):1021-39.
2. ICHD-3. *Cephalalgia.* 2018;38(1):1-211.

Disclosures

Dr. Charleston has received personal compensation for serving as a consultant for Allergan/AbbVie, Amgen, Amneal, Aurene, Biohaven, Haleon, LinPharma, Pfizer, and Satsuma; is on the advisory panel for Mi- Helper Inc. (stock options); has received grant/research support from the Disparities in Headache Advisory Council and Amgen; has received CME honoraria from American Headache Society, American Academy of Neurology, Migraine360 CME Program, and the Primary Care Education Consortium; receives a salary as faculty from Michigan State University College of Human Medicine and Thomas Jefferson University; is a non-compensated associate editor for *Headache: The Journal of Head and Face Pain*; and serves as a non-compensated Board of Directors as Treasurer/Secretary for the Clinical Neurological Society of America.

Dr. Strom is an employee of Satsuma Pharmaceuticals and was a stockholder at the time of study conduct.

Dr. Albrecht was an employee and stockholder of Satsuma Pharmaceuticals at the time of study conduct and is now a consultant and stockholder for Satsuma Pharmaceuticals.

Dr. Cowan is a co-founder of BonTriage, LLC and Agonex, Inc. and has received consulting fees from AbbVie, Biohaven, Eli Lilly, Nupathe, Satsuma, Teva, and Tonix.

Acknowledgements

This study and medical writing support were funded by Satsuma Pharmaceuticals, Inc. (Durham, NC, USA). Medical writing support was provided by The Medicine Group, LLC (New Hope, PA, USA) in accordance with Good Publication Practice guidelines.

Presented at the American Headache Society (AHS) Annual Meeting; June 13–16, 2024