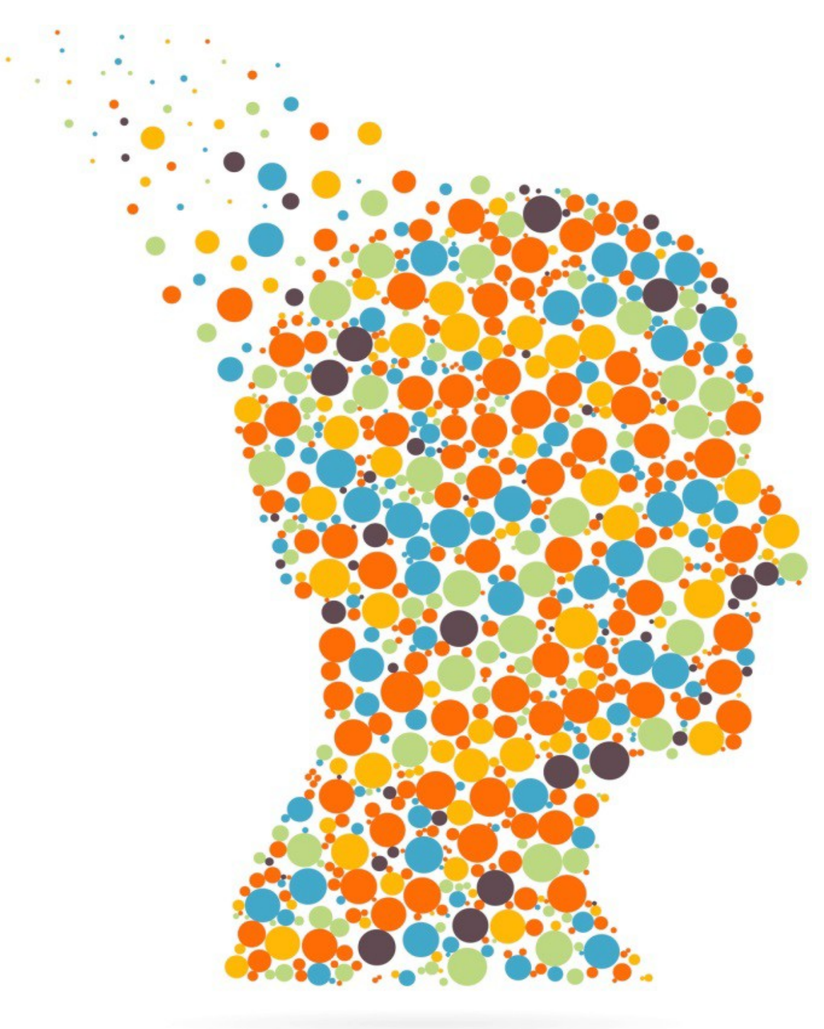

Comparison of the Pharmacokinetics of STS101, an Intranasal Dry Powder Formulation of Dihydroergotamine, with Other Intranasal, Injectable, and Oral Inhaled DHE Formulations

Shannon Strom, PhD[1]; John Kollins[1]; and Detlef Albrecht, MD[1]
[1]Satsuma Pharmaceuticals, Inc., South San Francisco, United States

IHC-PO-362

Objectives

To perform a literature review to compare pharmacokinetic (PK) parameters for several formulations of dihydroergotamine mesylate (DHE) including intravenous (IV), intramuscular (IM), intranasal liquid sprays (IN), orally inhaled pulmonary, and IN dry powder that are marketed, have demonstrated anti-migraine efficacy in controlled Phase 3 studies, or are in active development.

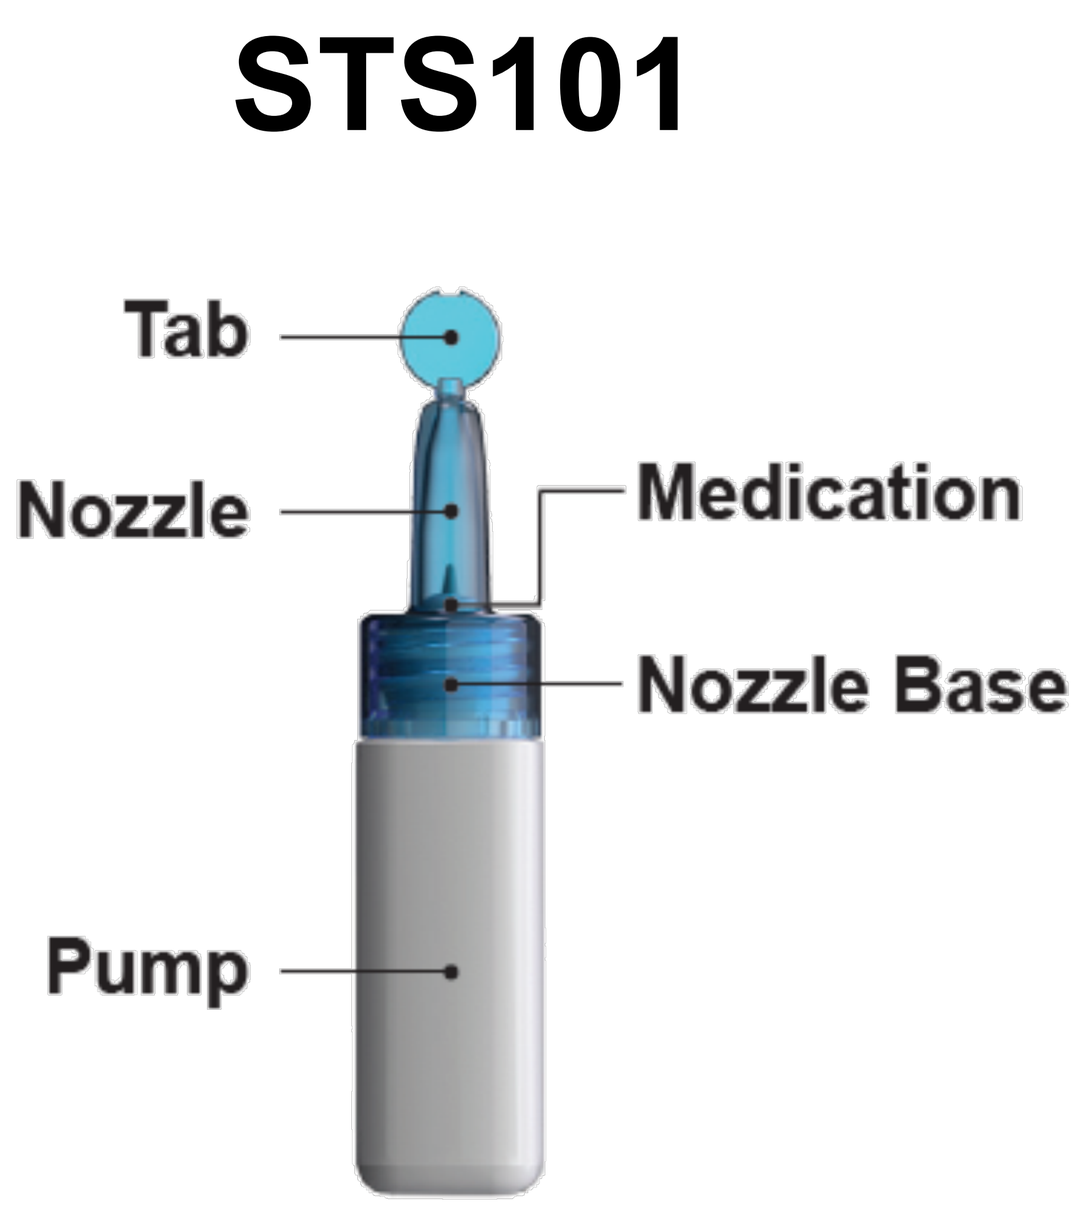
STS101
Tab
Nozzle
Medication
Nozzle Base
Pump

Methods

A literature search was performed to compare the PK results of several formulations of DHE including the approved products Migranal® (liquid nasal spray) and D.H.E. 45® (administered IV and IM), and the development-stage products INP104 (liquid nasal spray), MAP0004 (orally inhaled pulmonary whose development has been discontinued), and STS101 (dry powder nasal formulation). The 5.2 mg DHE dose for STS101 is equivalent to 6.0 mg DHE mesylate.

The data are presented from cross-trial comparisons, not head-to-head clinical studies. The study protocol designs, subject demographics, and analyses were similar across the studies so comparisons can be made.

One item that differed across the studies was the plasma DHE analytical assay, as more sensitive assays were developed over time. The analytical method used through 1996 was a radioimmunoassay with a limit of quantitation of 20 pg/mL. The more recent investigational development programs used high-performance liquid chromatography in conjunction with tandem mass spectrometry (LC-MS/MS) methods with a limit of quantitation of 10 pg/mL[5, 8] and 8 pg/mL[7].

Extraction of DHE plasma concentration curves from the published literature was accomplished using WebPlotDigitizer by Automeris.

If a coefficient of variation was not provided in the published literature, it was calculated as the ratio of the standard deviation to the mean.

The estimated minimum threshold concentration for efficacy is derived from the Migranal Summary Basis of Approval, 1997.

Results

- Rapid freedom from pain and associated symptoms are the goals for the acute treatment of migraine, so C_{max} and $AUC_{0\text{-}2hr}$ are important predictors of a clinical response[2, 3, 5, 9].
- STS101 achieved rapid and sustained high drug exposure with low variability (Table 1). STS101 had a higher C_{max} than intranasal liquid sprays Migranal and INP104 and approached that of the orally inhaled MAP0004.
- The STS101 $AUC_{0\text{-}2hr}$ was 2-fold or more greater than for Migranal, INP104, and MAP0004. The STS101 $AUC_{0\text{-}inf}$ was 2-fold or more greater than for Migranal, INP104, and MAP0004. STS101 achieved 83% of the total drug exposure ($AUC_{0\text{-}inf}$) of IM DHE and was comparable to IV DHE (Table 1).
- STS101 achieved higher cumulative drug exposure than Migranal, INP104, and MAP0004 by approximately 30 minutes and all time points thereafter (Figure 1).
- Plasma concentrations and AUC values for Migranal were similar across multiple historical studies and in the STS101 Phase 1 study, but were lower in the INP104 Phase 1 study (Figure 2, Table 1).
- The coefficient of variation (%CV) for C_{max}, $AUC_{0\text{-}2hr}$, and $AUC_{0\text{-}inf}$ were lower with STS101 versus Migranal and INP104 and were similar to MAP0004 (Figure 3).

Conclusions

- STS101 demonstrated higher PK (C_{max}, $AUC_{0\text{-}2hr}$, and $AUC_{0\text{-}24hr}$) and less PK variability (%CV) as compared to the DHE liquid nasal spray products (Migranal and INP104).
- As compared with orally inhaled pulmonary DHE (MAP0004), STS101 achieved similar C_{max} and higher AUC at all time points after ~30 minutes as well as similar PK variability (%CV).
- STS101 showed high sustained plasma concentrations and $AUC_{0\text{-}inf}$ comparable to the IV DHE values (but avoiding the high Cmax of IV DHE associated with adverse events) and approaching the IM DHE values.
- Lower PK variability may lead to more consistent and reliable clinical performance.
- The PK profile of STS101 predicts that the acute treatment goals of rapid and consistent pain freedom and associated symptoms without recurrence should be achieved[1].
- A Phase 3 efficacy study (EMERGE) with STS101 is ongoing to evaluate the product as an acute treatment for migraine (NCT 03901482).

Table 1. Pharmacokinetic Parameters for Different Routes of Administration of DHE

DHE Mesylate Formulation	Mean C_{max} ± SD (pg/mL) %CV	Median T_{max} [min, max] (hr)	Mean $AUC_{0\text{-}2hr}$ ± SD (pg*mL/hr) %CV	Mean $AUC_{0\text{-}inf}$ ± SD (pg*mL/hr) %CV
D.H.E. 45 IV (1 mg)				
Kellerman, 2013[1]	54,189 ± 34,970 (%CV: 64.5)	0.067 (0.000, 0.100)	7,331 ± 3,194 (%CV: 43.6)	12,894 ± 3,976 (%CV: 30.8)
Shrewsbury, 2019	14,620 ± 4,906 (%CV: 33.6)	0.08 (0.07, 0.10)	3,019 ± 513.4 (%CV: 17.0%[2])	7,381 ± 1,139 (%CV: 15.4)
D.H.E. 45 IM (1 mg)				
Humbert, 1996	4,440 ± 1,250 (%CV = 28%[2])	0.38 ± 0.30 hours	Not Provided	14,490 ± 3,780 (%CV = 26%[2])
STS101-001 Phase 1 Study	3,368 ± 840 (%CV: 24.9)	0.25 (0.08, 1.00)	4,791 ± 907 (%CV: 18.9)	13,650 ± 2,143 (%CV: 15.7)
Migranal Nasal Spray (1 mg)				
Humbert, 1996	1,020 ± 420 (%CV = 41%[2])	0.93 ± 0.63 hours	Not Provided	5,320 ± 2,300 (%CV = 43%[2])
Migranal Nasal Spray (2 mg)				
Migranal SBA Study 303-022[3]	1,131 ± 502[4] (%CV = 44.4[2])	0.878 (not provided)	Not Provided	Not Provided
STS101-001 Phase 1 Study	960 ± 726 (%CV: 75.7)	1.00 (0.50, 2.00)	1,316 ± 989 (%CV: 75.2)	6,498 ± 3,551 (%CV: 54.7)
Shrewsbury, 2019	329 ± 261 (%CV: 79.4)	0.67 (0.50, 1.8)	428.7 ± 317 (%CV: 74.1[2])	2,208 ± 1,488 (%CV: 67.4)
MAP0004 Orally Inhaled (1 mg)				
Kellerman, 2013[1]	2,720 ± 1,088 (%CV: 40.0)	0.167 (0.067, 0.25)	1,447 ± 541 (%CV: 37.4)	4,472 ± 1,530 (%CV: 34.2%)
INP104 Intranasal Spray (1.45 mg)	1,281 ± 682 (%CV: 53.3)	0.50 (0.33, 0.78)	1,595 ± 800.9 (%CV: 50.2[2])	6,153 ± 2,721 (%CV: 44.2)
Shrewsbury, 2019				
STS101 Intranasal Powder (5.2 mg)				
STS101-001 Phase 1 Study	2,175 ± 884 (%CV: 40.7)	0.50 (0.25, 2.00)	2,979 ± 1,147 (%CV: 38.5)	12,030 ± 4,716 (%CV: 39.2)

[1] Data is from pooled analyses of 3 studies.
[2] %CV was not provided in the publication, so was calculated as the SD/mean.
[3] Study 303-022 from the Migranal Summary Basis of Approval, 1997
[4] The C_{max} data ranges from 1.13 ± 0.50 to 1.31 ± 0.27 across 4 PK studies in healthy volunteers presented in the Migranal Summary Basis of Approval, 1997.

Figure 1. STS101 DHE Plasma Concentrations Versus Other Non-Injected DHE Products

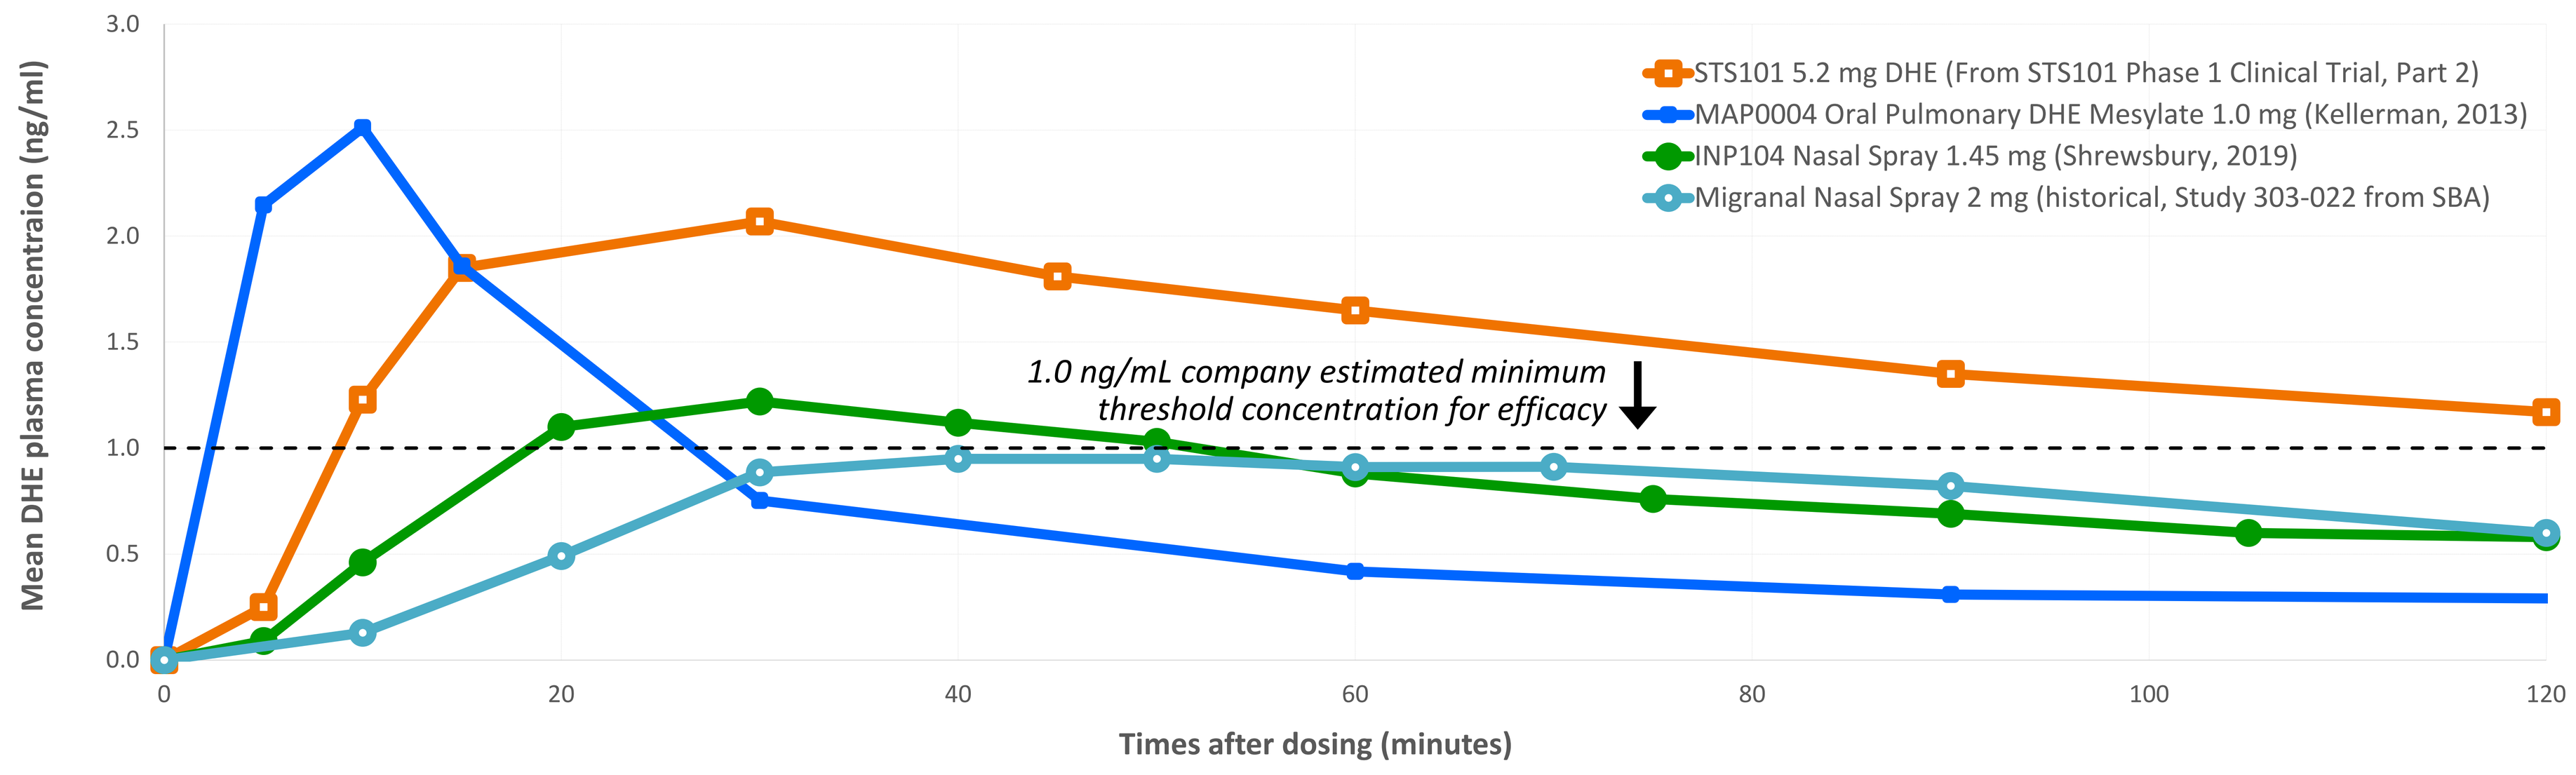
STS101 5.2 mg DHE (From STS101 Phase 1 Clinical Trial, Part 2)
MAP0004 Oral Pulmonary DHE Mesylate 1.0 mg (Kellerman, 2013)
INP104 Nasal Spray 1.45 mg (Shrewsbury, 2019)
Migranal Nasal Spray 2 mg (historical, Study 303-022 from SBA)
1.0 ng/mL company estimated minimum threshold concentration for efficacy
Mean DHE plasma concentraion (ng/ml)
Times after dosing (minutes)
0.0
0.5
1.0
1.5
2.0
2.5
3.0
0
20
40
60
80
100
120

Figure 2. Plasma Concentrations for DHE Liquid Nasal Sprays are Similar Across Multiple Studies Except the INP104 Phase 1 Study

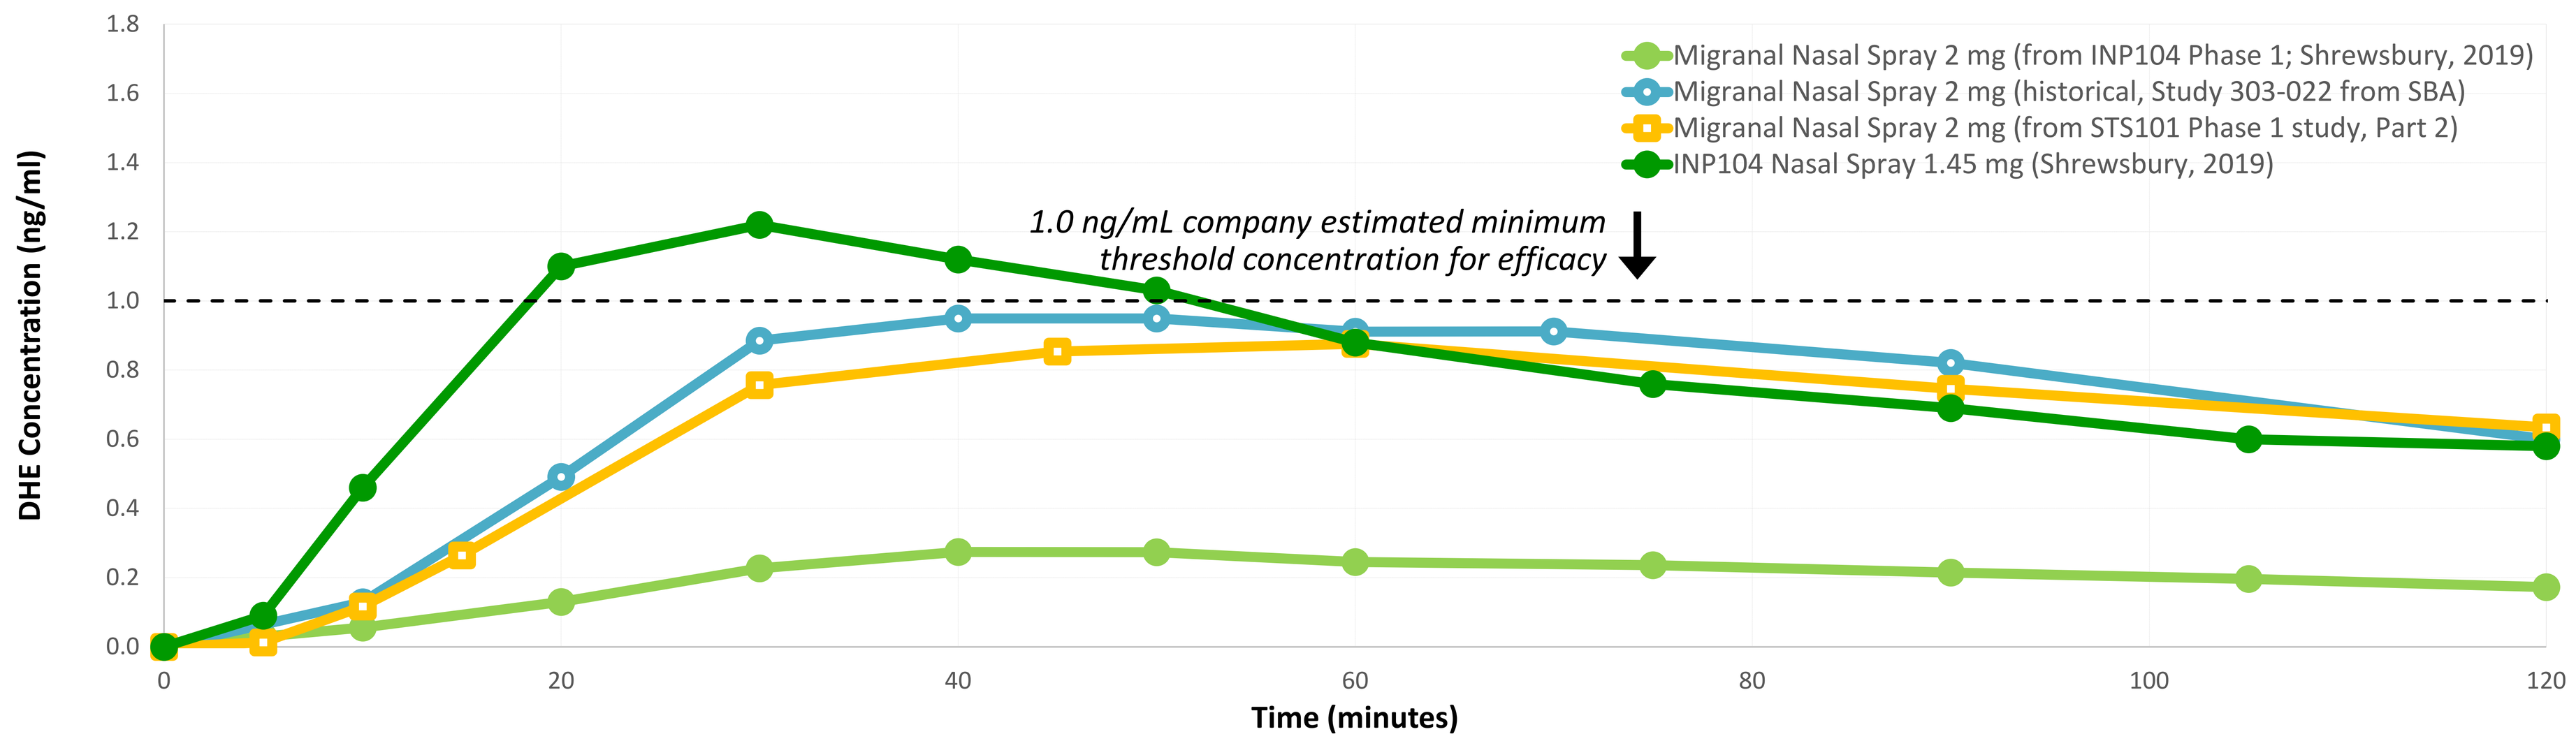
Migranal Nasal Spray 2 mg (from INP104 Phase 1; Shrewsbury, 2019)
Migranal Nasal Spray 2 mg (historical, Study 303-022 from SBA)
Migranal Nasal Spray 2 mg (from STS101 Phase 1 study, Part 2)
INP104 Nasal Spray 1.45 mg (Shrewsbury, 2019)
1.0 ng/mL company estimated minimum threshold concentration for efficacy
DHE Concentration (ng/ml)
Time (minutes)
0.0
0.2
0.4
0.6
0.8
1.0
1.2
1.4
1.6
1.8
0
20
40
60
80
100
120

Figure 3. STS101 C_{max} and AUC Coefficient of Variation for STS101 are Lower than DHE Liquid Nasal Sprays and Comparable to Oral Pulmonary DHE in a Cross-Trial Comparison

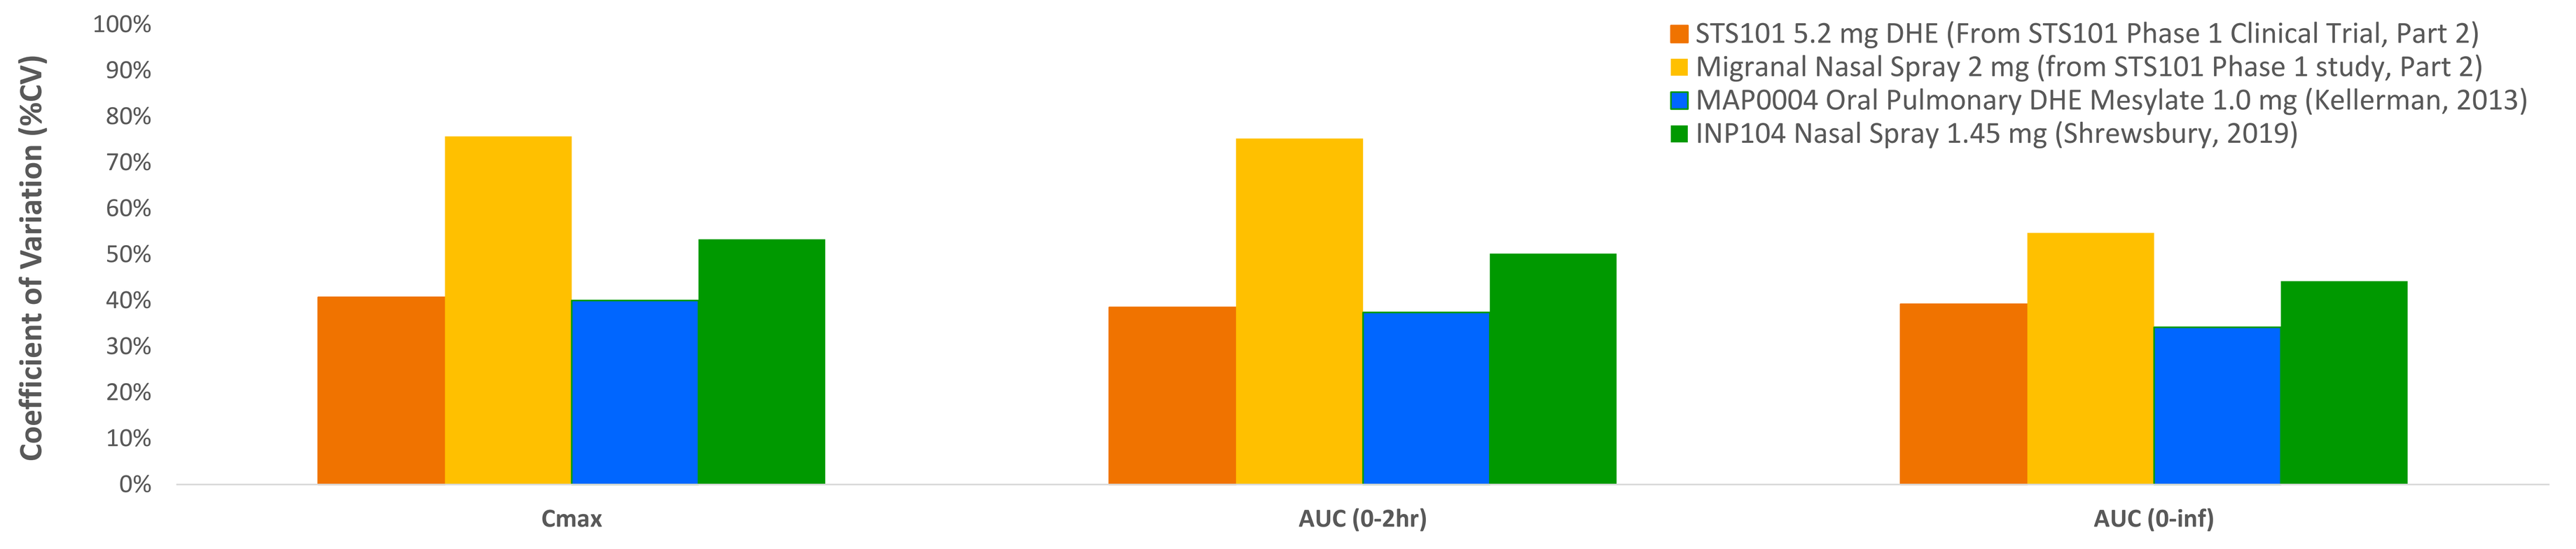
STS101 5.2 mg DHE (From STS101 Phase 1 Clinical Trial, Part 2)
Migranal Nasal Spray 2 mg (from STS101 Phase 1 study, Part 2)
MAP0004 Oral Pulmonary DHE Mesylate 1.0 mg (Kellerman, 2013)
INP104 Nasal Spray 1.45 mg (Shrewsbury, 2019)
Coefficient of Variation (%CV)
0%
10%
20%
30%
40%
50%
60%
70%
80%
90%
100%
Cmax
AUC (0-2hr)
AUC (0-inf)

Take a picture to download

References

1. American Headache Society Consensus Statement. The American Headache Society Position Statemen on Integrating New Migraine Treatments into Clinical Practice. Headache. 0:1-18, 2018.
2. FDA Guidance for Industry: Migraine: Developing Drugs for Acute Treatment, February 2018.
3. Ferrari A, Pinetti D, Bertolini A, Coccia C, Sternieri E. Interindividual Variability of Oral Sumatriptan Pharmacokinetics and of Clinical Response in Migraine Patients. Eur J Clin Pharmacol. 64:489-495, 2008.
4. Humbert H, Cabiac MD, Dubray C, and Lavene D. Human Pharmacokinetics of Dihydroergotamine Administered by Nasal Spray. Clin Pharmacol Ther 60:265-275, 1996.
5. Kellerman DJ, Forst A, Combs DL, Borland S, Kori S. Assessment of the Consistency of Absorption of Dihydroergotamine Following Oral Inhalation: Pooled Results from Four Clinical Studies. J. Aerosol Medicine and Pulmonary Drug Delivery. 26(5):297-306, 2013.
6. Migranal® US Food and Drug Administration, Summary Basis of Approval, 1997.
7. Satsuma Pharmaceuticals Study STS101-001 Phase 1 Clinical Study Report, May 2019.
8. Shrewsbury SB, Jeleva M, Satterly K, Lickliter J, Hoekman J. STOP 101: A Phase 1, Randomized, Open-Label, Comparative Bioavailability Study of INP104, Dihydroergotamine Mesylate (DHE) Administered Intranasally by a I123 Precision Olfactory Delivery (POD®) Device, in Healthy Adult Subjects. Headache, 0:1-16, 2019.
9. Thomsen LL, Dixon R, Lassen LH, Giboens M, Langemark M, Bendtsen L, Daugaard D, Olesen J. 311C90 (Zolmitriptan), a Novel Centrally and Peripheral Acting Oral 5-Hydroxytryptamine-1D Agonist: A Comparison of its Absorption During a Migraine Attack and in a Migraine-Free Period. Cephalalgia, 16:270-5, 1996.

Presented at the International Headache Conference, 5-8 September 2019