STS101 (Dihydroergotamine Nasal Powder) Shows Long Duration Anti-Migraine Benefit on Baseline Photophobia, Phonophobia, and Nausea: Results From the Phase 3 Double-Blind, Randomized, Placebo-Controlled SUMMIT Study

P-611

Christopher Gottschalk, MD[1], Egilius L.H. Spierings, MD[2], Shannon Strom, PhD[3], Detlef Albrecht, MD[3]

[1]Yale University, New Haven, CT, USA; [2]MedVadis Research, Waltham, MA, USA; [3]Satsuma Pharmaceuticals, Inc., Durham, NC, USA

Introduction

- Dihydroergotamine mesylate (DHE) is a recommended first-line treatment option for the acute treatment of moderate or severe migraine attacks, with or without aura.[1]
- STS101 is a novel investigational DHE product that combines a mucoadhesive nasal powder formulation delivered with an easy-to-use, easy-to-carry, pre-filled, single-use nasal delivery device (Figure 1).
- The STS101 advanced nasal powder and device technology maximizes deposition of DHE on the nasal mucosa, enhancing DHE absorption, increasing drug exposure and reducing pharmacokinetic variability in comparison with DHE liquid nasal sprays.[2]
- STS101 is the only DHE-containing product with double-blind, placebo-controlled phase 3 efficacy data using current state of the art clinical trial endpoints recommended by the FDA[3] and the International Headache Society.[4]

Objective

- To compare the cardinal symptom reduction capabilities of STS101 compared with placebo in the acute treatment of migraine attacks in a double-blind, randomized, placebo-controlled phase 3 study (SUMMIT)

Methods

Study Design and Treatment Intervention

- SUMMIT was a double-blind, randomized, placebo-controlled phase 3 trial in adults with a history of 2–8 moderate or severe migraine attacks and <15 headache days per month (NCT04940390).
- After establishing eligibility, the study participants self-administered a single 5.2-mg dose of STS101 or placebo to treat one migraine attack of moderate or severe pain intensity.

Participants

- Study participants must have had ≥1-year history of migraine (with or without aura) according to the International Classification of Headache Disorders, 3rd edition,[5] including:
 - Migraine onset before the age of 50 years
 - 2–8 migraine attacks/month with moderate or severe pain each month
 - <15 headache days/month
- Those with a non-migraine headache diagnosis, history of cerebrovascular disease, ≥2 cardiovascular risk factors (hypertension, hypercholesteremia, obesity, diabetes mellitus, history of premature coronary artery disease, postmenopausal females, or males >46 years of age), and current (at screening) use of >1 migraine prevention treatment were excluded.

Outcomes and Analyses

- The cardinal symptoms of nausea, photophobia, and phonophobia were measured at baseline immediately before treatment of the migraine headache and up to 48 hours after treatment.
 - Freedom from nausea, phonophobia, or photophobia were defined as absence of nausea, phonophobia, or photophobia post-dose if present at baseline
- Efficacy data are presented for the modified intent-to-treat (mITT) population, defined as all randomized participants who reported a qualifying migraine attack, received the study medication, and had reported efficacy data in ≥1 post-treatment eDiary entry through an analysis that treats missing data as a non-responder.
- Participants who did not have evaluable assessments at the 2-hour time point, or who received rescue medication prior to the 2-hour time point, were considered non-responders.

Results

- Of 1,591 randomized participants, 1,424 were evaluable for efficacy analysis (Table 1).
- The treated attacks showed high rates of severe pain (38%), nausea (69%), photophobia (96%), phonophobia (91%), and allodynia (63%) (Table 2).
- A higher proportion of participants (n=324/496) who experienced nausea pre-dose reported freedom from nausea starting at 3 hours post-dose with STS101 compared with placebo (n=272/483), demonstrating nominal statistical significance through 48 hours post-dose (Figure 2).
- A higher proportion of participants (n=324/683) who experienced photophobia pre-dose reported freedom from photophobia compared with placebo (n=279/687) starting at 3 hours post-dose, demonstrating nominal statistical significance through 48 hours post-dose (Figure 3).
- More participants with phonophobia pre-dose (n=347/648) reported freedom from phonophobia starting at 3 hours post-dose with STS101 compared with placebo (n=268/642), through 48 hours post-dose (Figure 4).

Figure 1. STS101 Administration

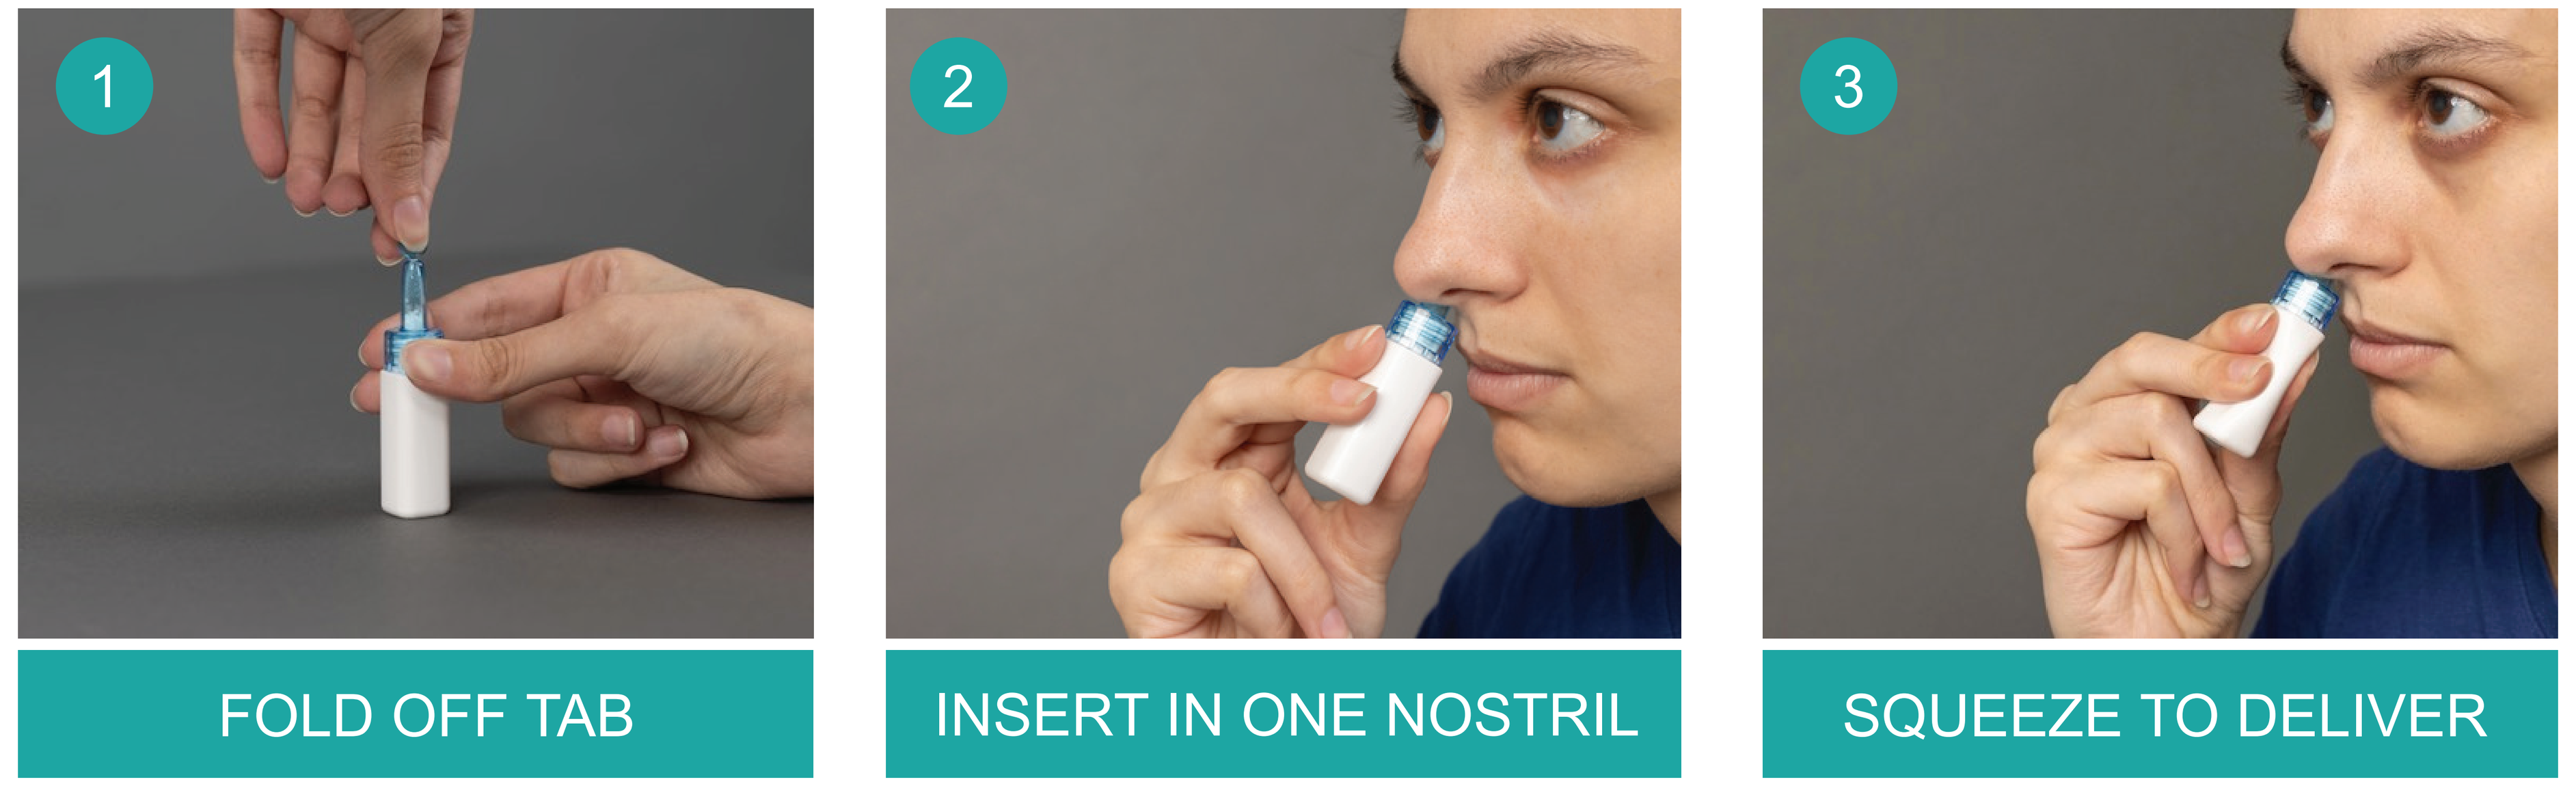
1
2
3
FOLD OFF TAB
INSERT IN ONE NOSTRIL
SQUEEZE TO DELIVER

Table 1. Demographics and Baseline Characteristics (mITT)

	STS101 5.2 mg n=716	Placebo n=708
Age, mean (SD)	38.2 (11.4)	38.9 (11.5)
Sex, % male/female	21/79	20/80
Race, %		
White	82.5	81.4
African American	12.0	13.7
Asian	3.5	3.4
Other	1.0	0.7
Years since onset, mean (SD)	16.2 (11.3)	16.9 (1.7)
Monthly migraines & headache days reported before screening, mean (SD)		
Moderate or severe	4.5 (1.7)	4.5 (1.7)
Headache days	6.9 (2.7)	6.9 (3.0)
Participants on migraine prevention medication at randomization, %	10.2	10.2
Mean HIT-6 score (% of participants with severe impact [>60])	64.2 (88.1)	64.1 (86.4)

Data are for the modified intent-to-treat population. HIT-6, 6-item Headache Impact Test; SD, standard deviation.

Table 2. Migraine Attack Baseline Symptoms

	STS101 5.2 mg n=716	Placebo n=708
Migraine headache severity at baseline, n (%)		
Moderate	441 (61.6)	449 (63.4)
Severe	275 (38.4)	259 (36.6)
Allodynia at baseline, n (%)	450 (62.8)	444 (62.7)
Nausea at baseline, n (%)	496 (69.3)	483 (68.2)
Photophobia at baseline, n (%)	683 (95.4)	687 (97.0)
Phonophobia at baseline, n (%)	648 (90.5)	642 (90.7)
Functional impairment status, n (%)		
Moderate	392 (54.7)	365 (51.6)
Severe	205 (28.6)	208 (29.4)

Figure 2. Proportion of Participants With Freedom From Nausea Through 48 Hours Post-Dose

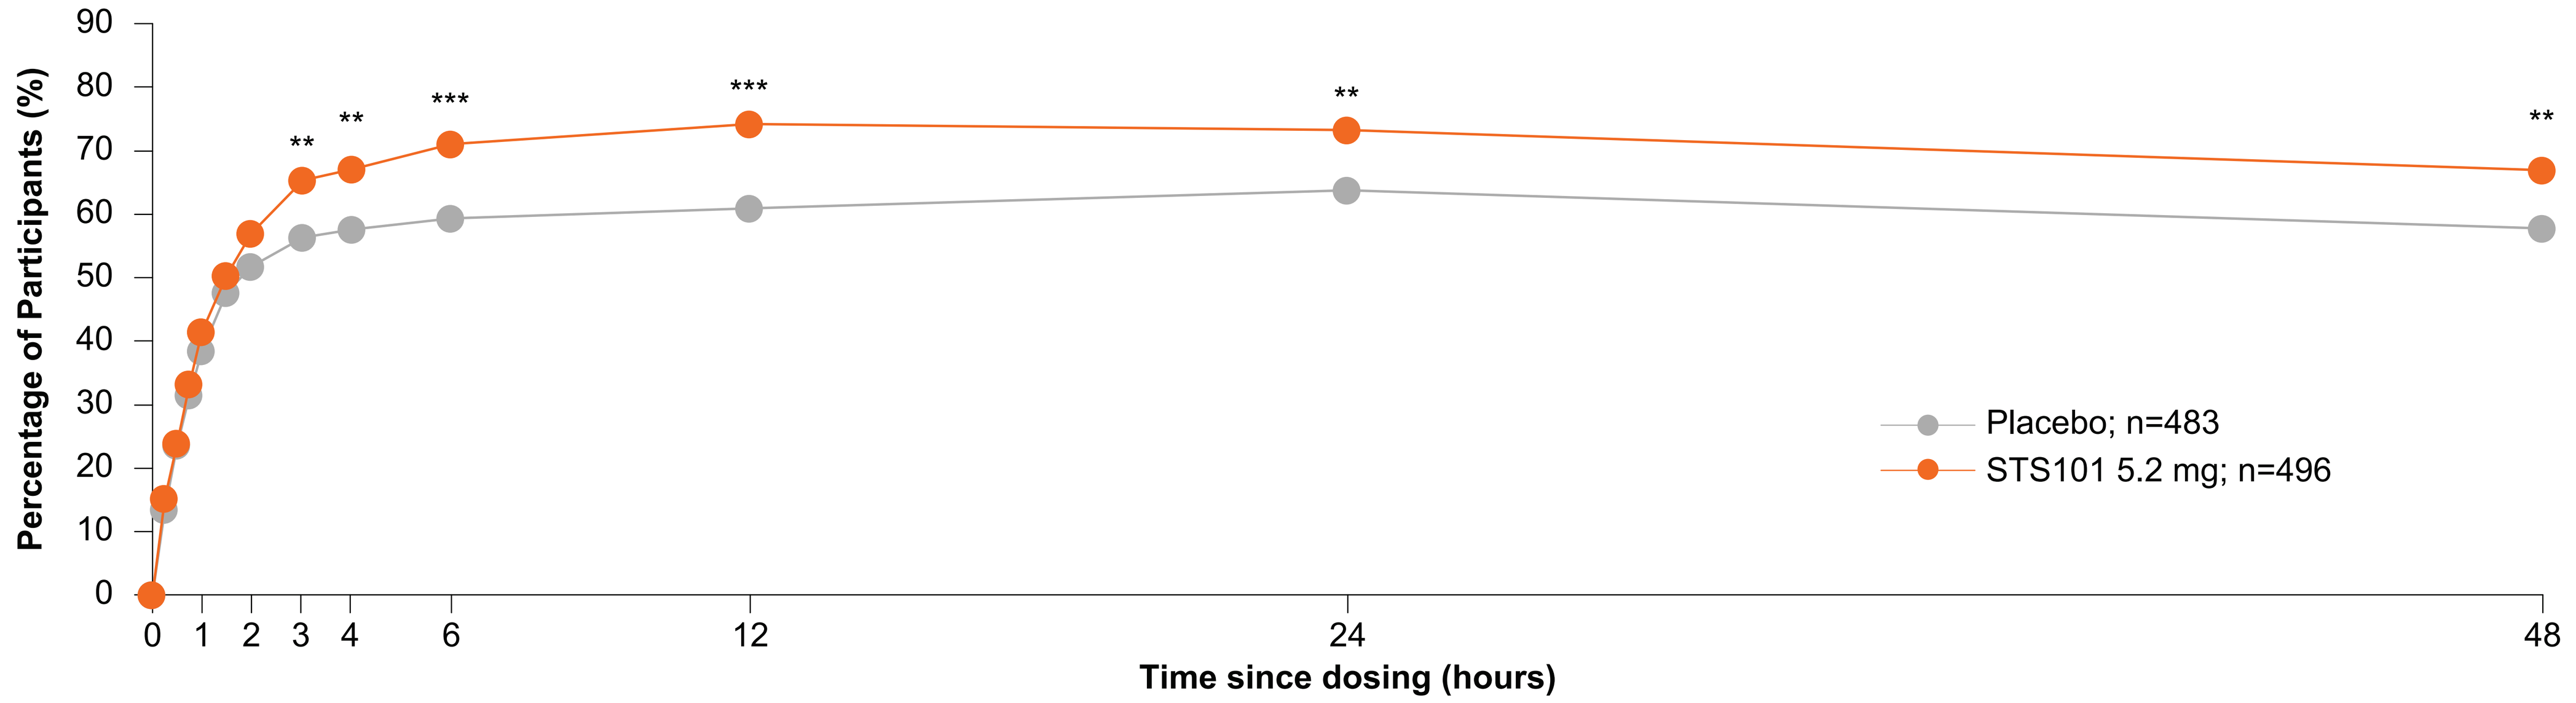
Percentage of Participants (%)
Time since dosing (hours)
Placebo; n=483
STS101 5.2 mg; n=496

p<0.01, *p<0.001

Figure 3. Proportion of Participants With Freedom From Photophobia Through 48 Hours Post-Dose

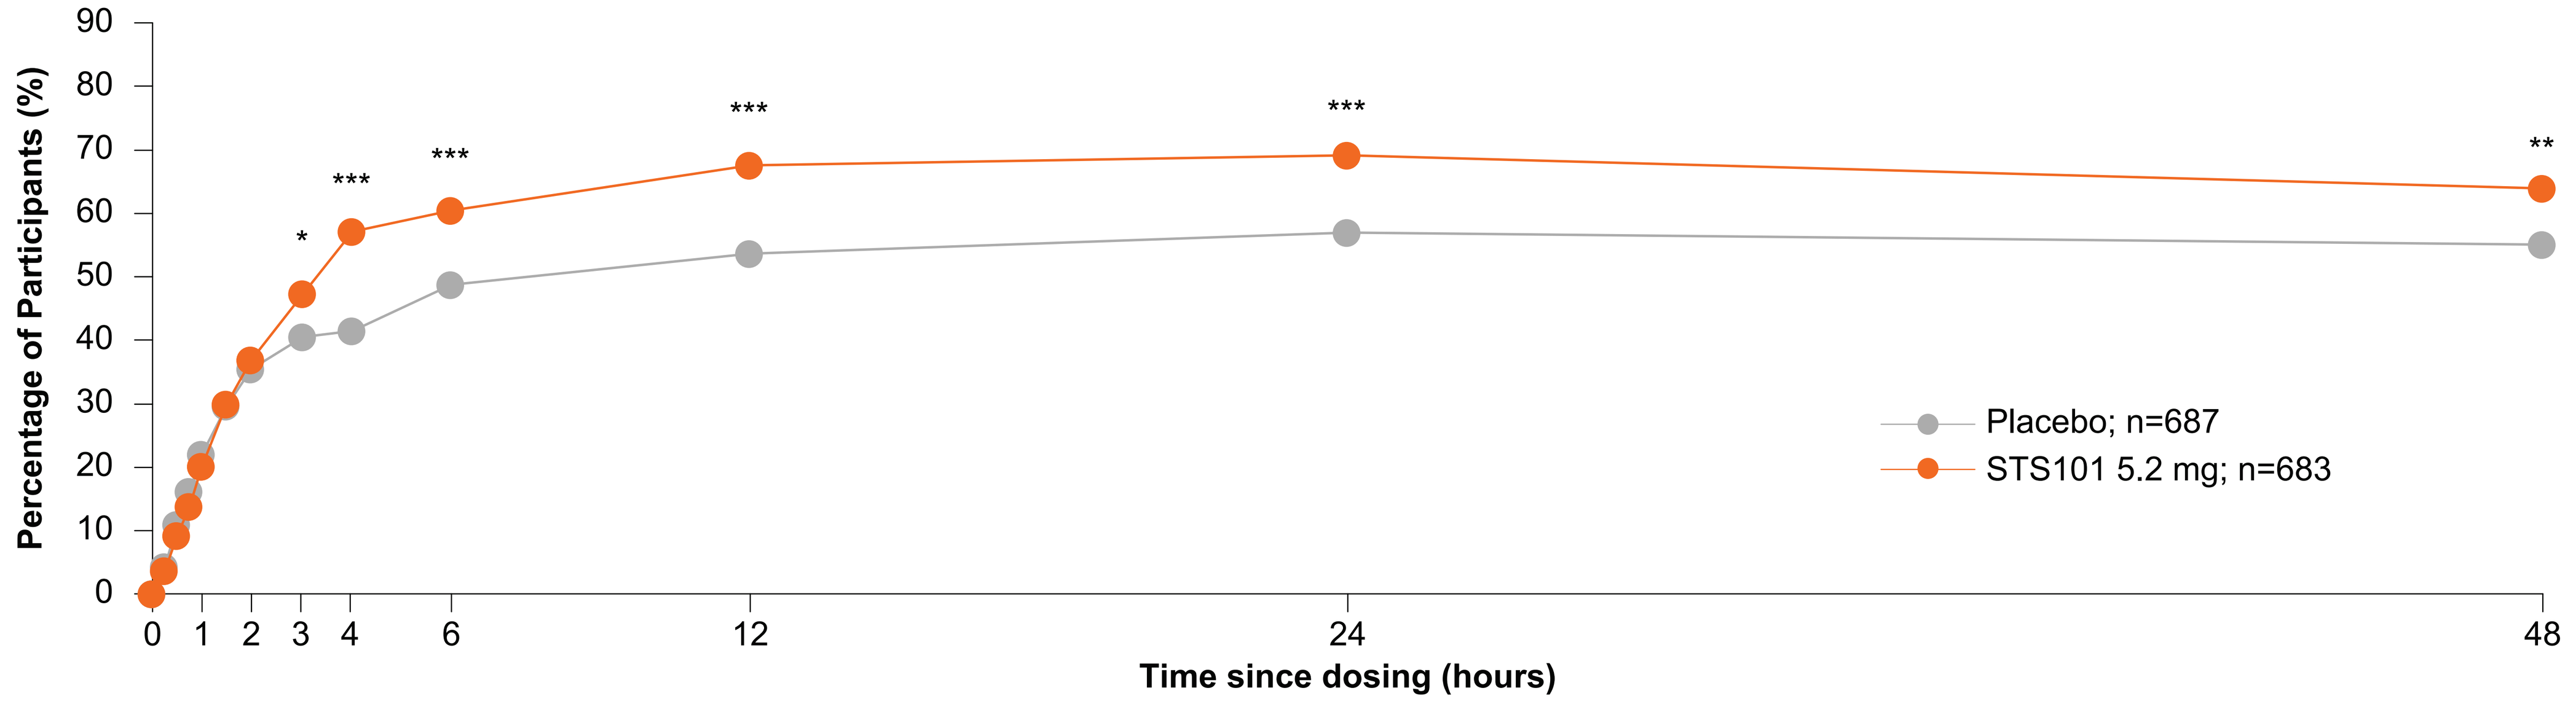
Percentage of Participants (%)
Time since dosing (hours)
Placebo; n=687
STS101 5.2 mg; n=683

*p<0.05, **p<0.01, ***p<0.001

Figure 4. Proportion of Participants With Freedom From Phonophobia Through 48 Hours Post-Dose

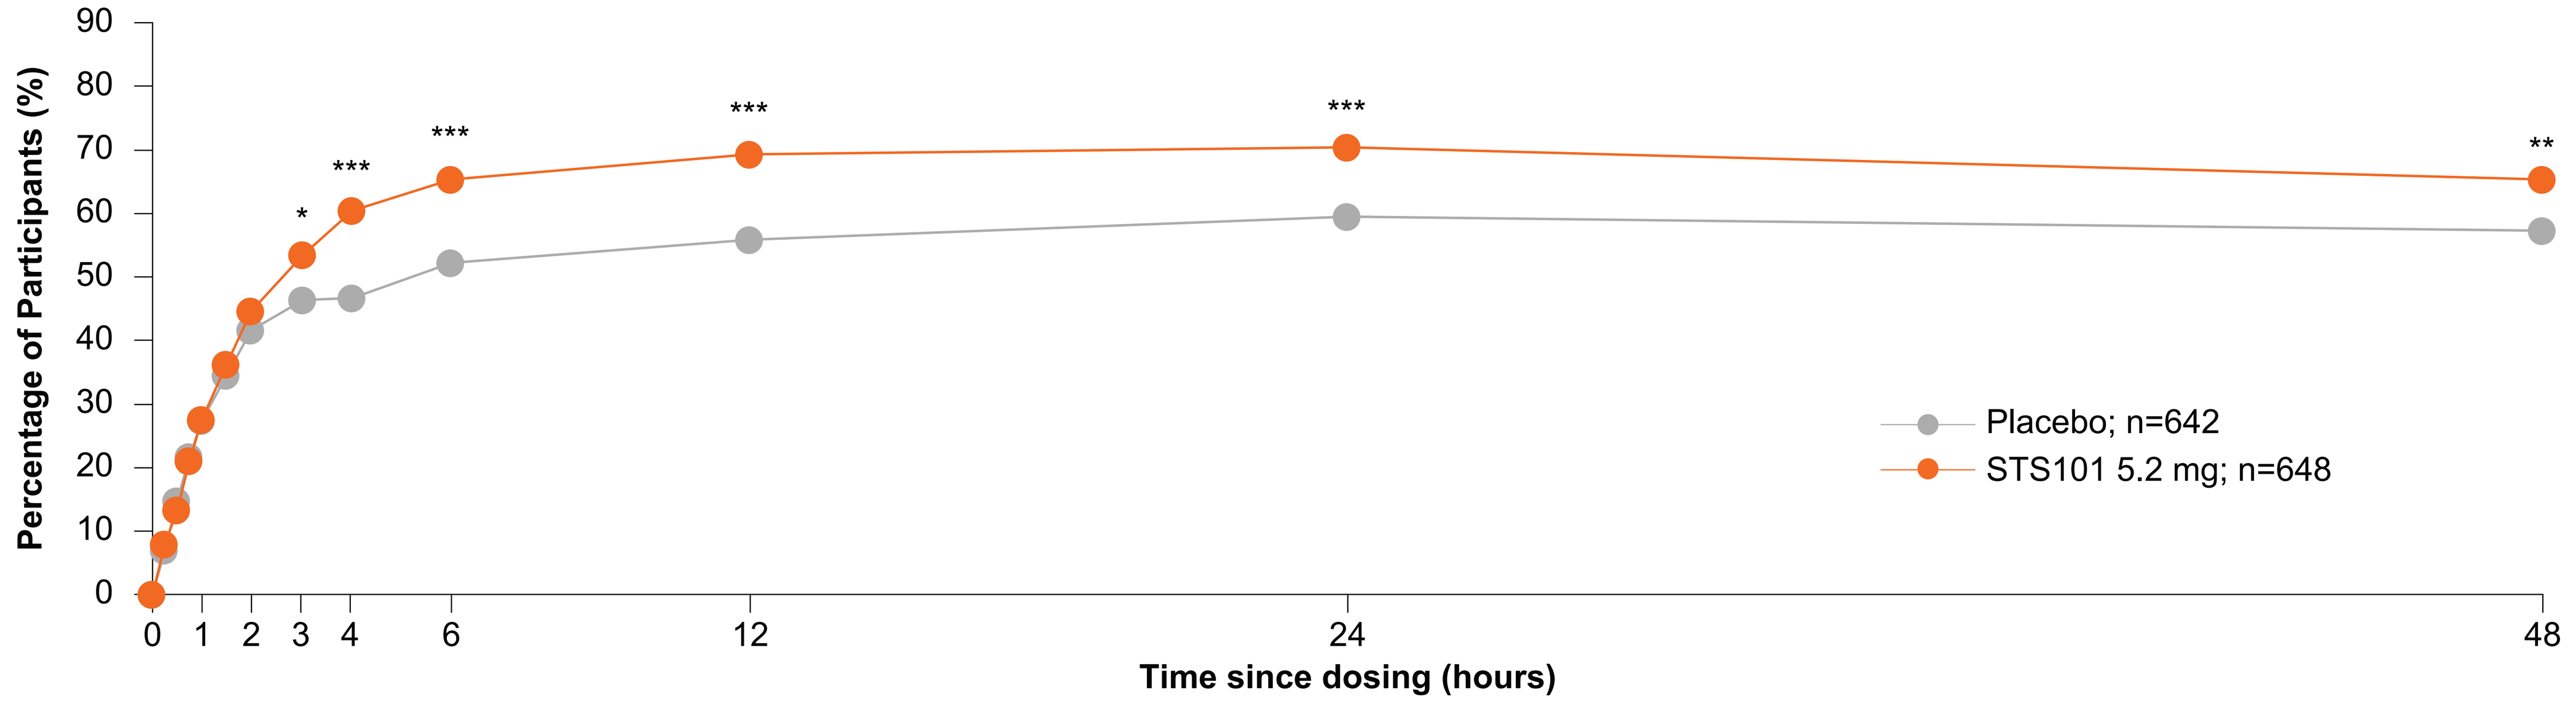
Percentage of Participants (%)
Time since dosing (hours)
Placebo; n=642
STS101 5.2 mg; n=648

*p<0.05, **p<0.01, ***p<0.001.

Conclusions

- The results of the SUMMIT study show that, similar to its effects on migraine headache pain, STS101 had significant anti-migraine effects versus placebo on the cardinal migraine symptoms of photophobia, phonophobia, and nausea from 3 through 48 hours post-dose.
- Specifically, the effect on nausea is noteworthy as other routes of DHE administration have been related to an increase in nausea.
- Lastly, STS101 proved to be effective in aborting the cardinal migraine symptoms in participants with difficult-to-treat attacks as indicated by high rates of allodynia, moderate and severe functional impact, and nausea among the baseline symptoms of the treated attacks.

References

1. Ailani J, et al. *Headache.* 2021;61(7):1021-39.
2. Lipton R, et al. *Headache.* 2024;64(3):266-75.
3. FDA Acute Migraine Guidance, 2018.
4. Diener HC, et al. *Cephalalgia.* 2019;39(6):687-710.
5. ICHD-3. *Cephalalgia.* 2018;38(1):1-211.

Disclosures

Dr. Gottschalk is a member of the scientific advisory boards for AbbVie, Lundbeck, Satsuma Pharmaceuticals, Theranica; a consultant for Spherix Global Insights; and a trustee of the Headache Cooperative of New England (HCNE).
Dr. Spierings is on the speakers bureau of AbbVie.
Dr. Strom is an employee of Satsuma Pharmaceuticals and was a stockholder at the time of study conduct.
Dr. Albrecht was an employee and stockholder of Satsuma Pharmaceuticals at the time of study conduct and is now a consultant for and stockholder for Satsuma Pharmaceuticals.

Acknowledgements

This study and publication were funded by Satsuma Pharmaceuticals (Durham, NC). The authors thank The Medicine Group, LLC (New Hope, PA, USA) for providing medical writing support, which was funded by Satsuma Pharmaceuticals, Inc. and in accordance with Good Publication Practice guidelines.

Presented at the American Headache Society (AHS) Annual Meeting; June 13–16, 2024